2605/8 - STX-1, a potent first-in-class ADC directed against DPP4 (CD26) as a biomarker of senescent tumor cells

Eric Angevin[1], Virginie Lelarge[2], Remi Capelle[2], Justine Choeur[2], Thierry Mathieu[2], Benjamin Le Calvé[2]

[1]Gustave Roussy, Villejuif, France, [2]StarkAge Therapeutics, Lille, France

Abstract

- Dipeptidyl Peptidase 4 (DPP4/CD26) is a membrane glycoprotein frequently expressed on malignant cells and recently identified as a novel biomarker of senescence[1].
- Many cancer therapies (i.e. chemo and radiation regimens or other drugs) induce a senescent phenotype (called 'SASP') with overexpression of DPP4 leading to the local production of pro-inflammatory cytokines, immunosuppressive factors and metalloproteinases that may promote tumor growth, invasion and neoangiogenesis[2,3].
- A proprietary[4] high-affinity anti-DPP4 IgG1κ mAb was generated as a candidate for cancer cells targeting and DPP4-mAb complexes are efficiently internalized. This mAb also results in *in vitro* inhibition of DPP4-positive human tumor cell lines induced to senescence by chemotherapies or Radiotherapy.
- STX-1 is a leading *first-in-class* ADC using the anti-DPP4 mAb. The first formulations of STX-1 evaluated preclinically were designed using a classical cleavable linker with two different payloads (Exatecan and MMAE) and were assessed for antitumor *in vitro* and *in vivo* activities.
- Exploiting the targeting of senescent cells in cancer via selected biomarkers such as DPP4 constitutes a new therapeutic avenue to counteract tumor dormancy, recurrences and resistance to conventional cancer therapies.

1. Rational for DPP4 targeting with STX-1

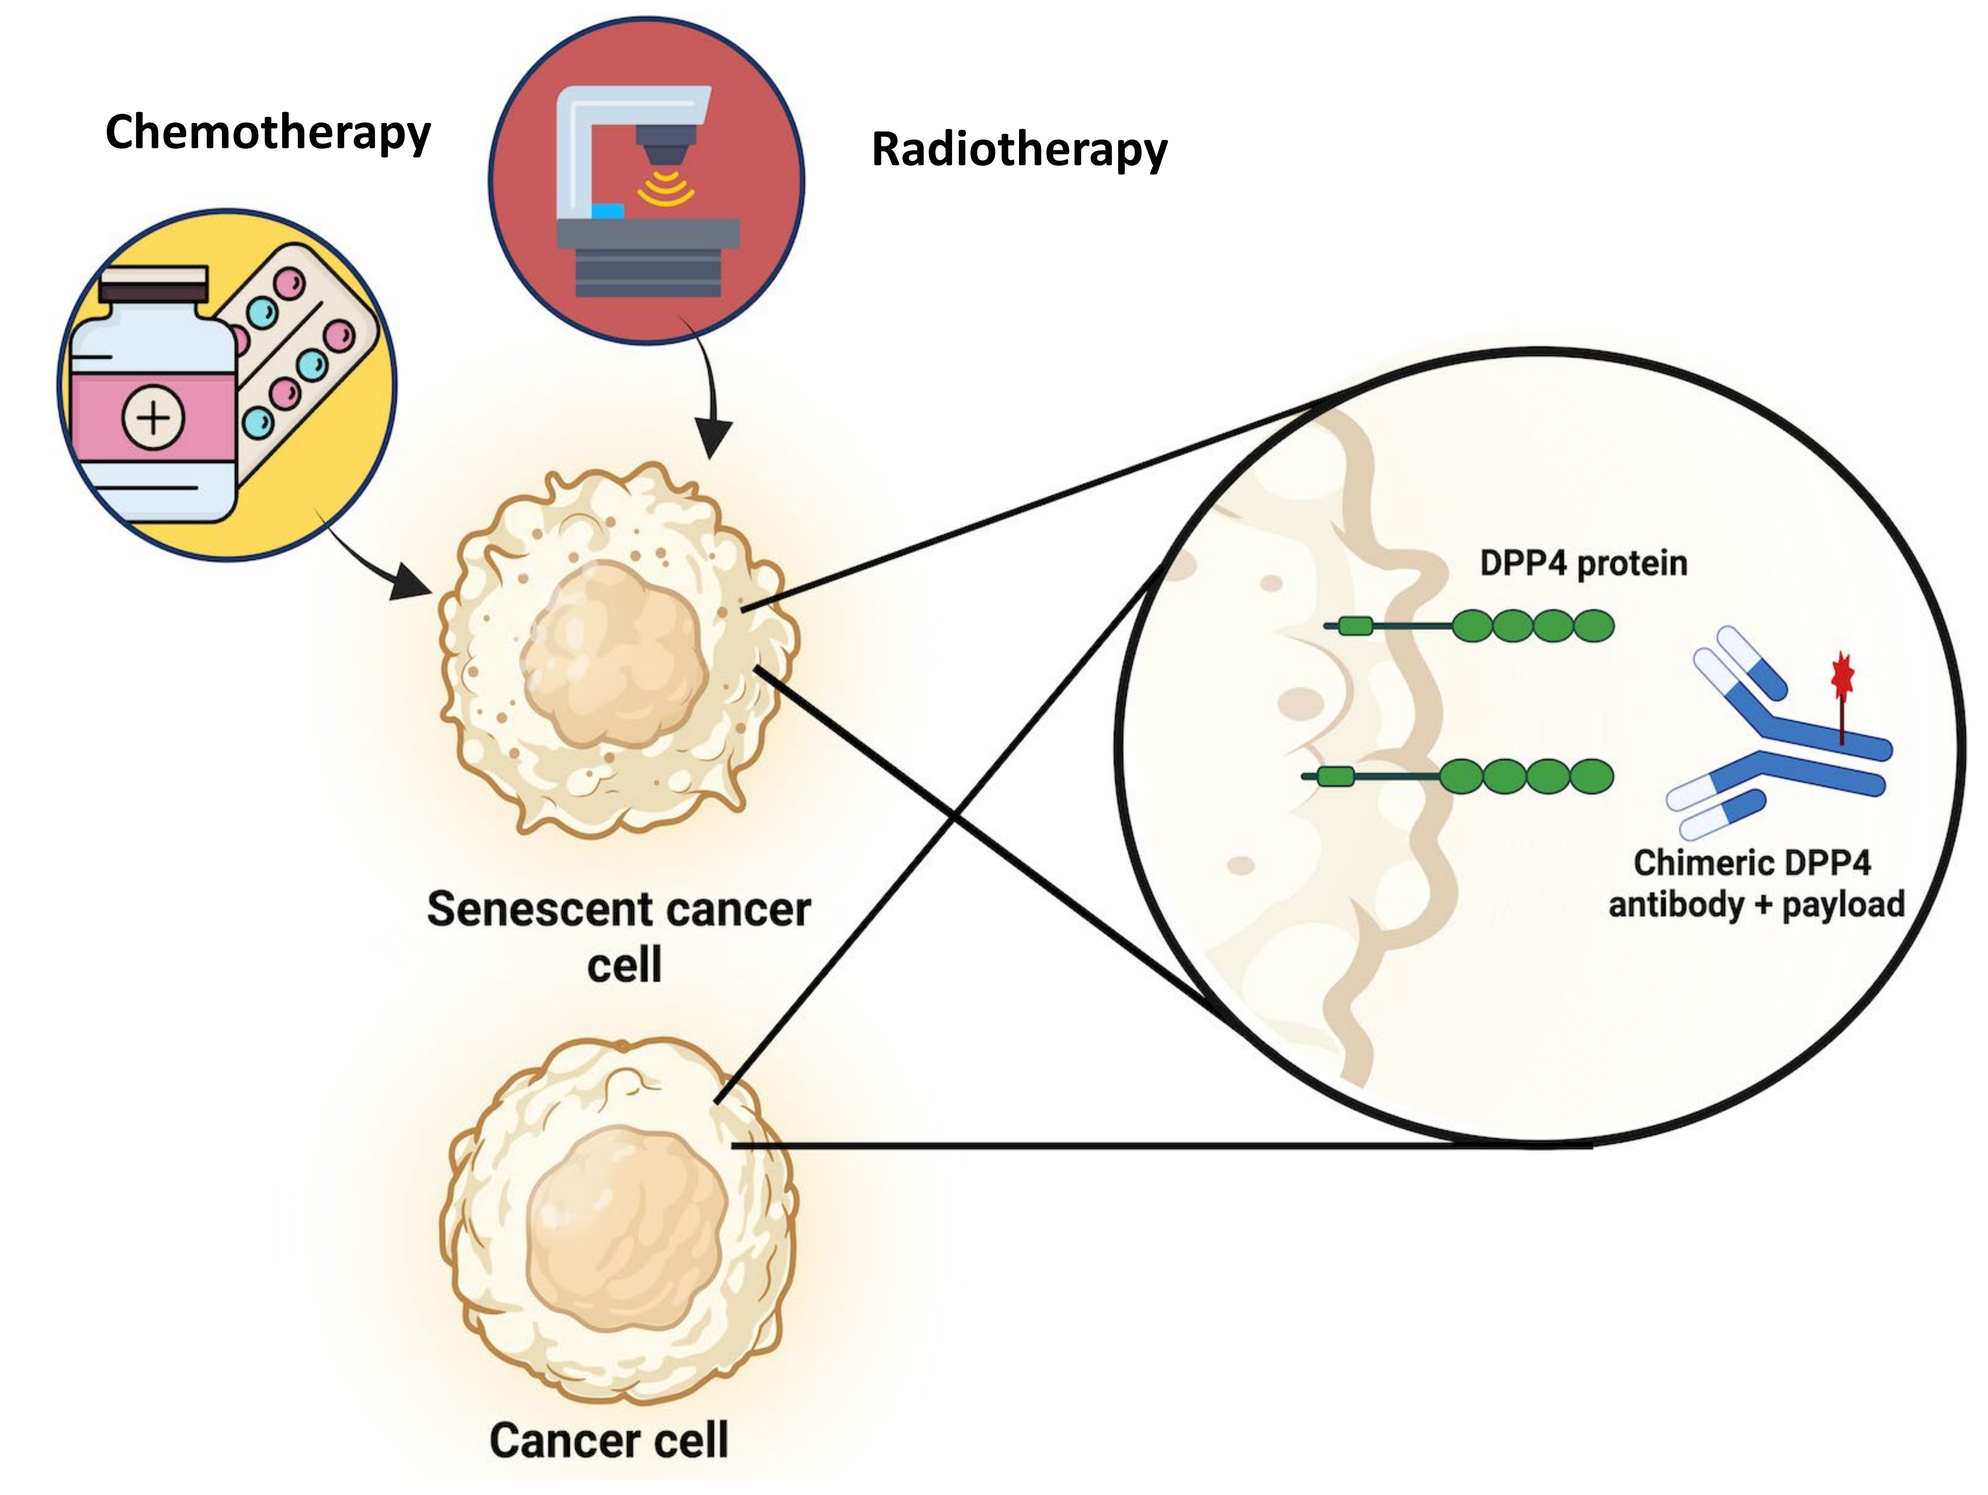

Chemotherapy
Radiotherapy
DPP4 protein
Chimeric DPP4 antibody + payload
Senescent cancer cell
Cancer cell

Fig.1 DPP4 (CD26) expression is boosted by senescence induction associated to drug resistance and tumor relapse after failure of conventional cancer therapies. Targeting DPP4 with an ADC is leading to the internalization of the complex into the endolysosomal compartments and to the release of the payload. STX-1 is the first generation of ADCs specifically designed to eliminate senescent cancer cells.

Results

2. DPP4 is expressed in a wide range of solid cancers

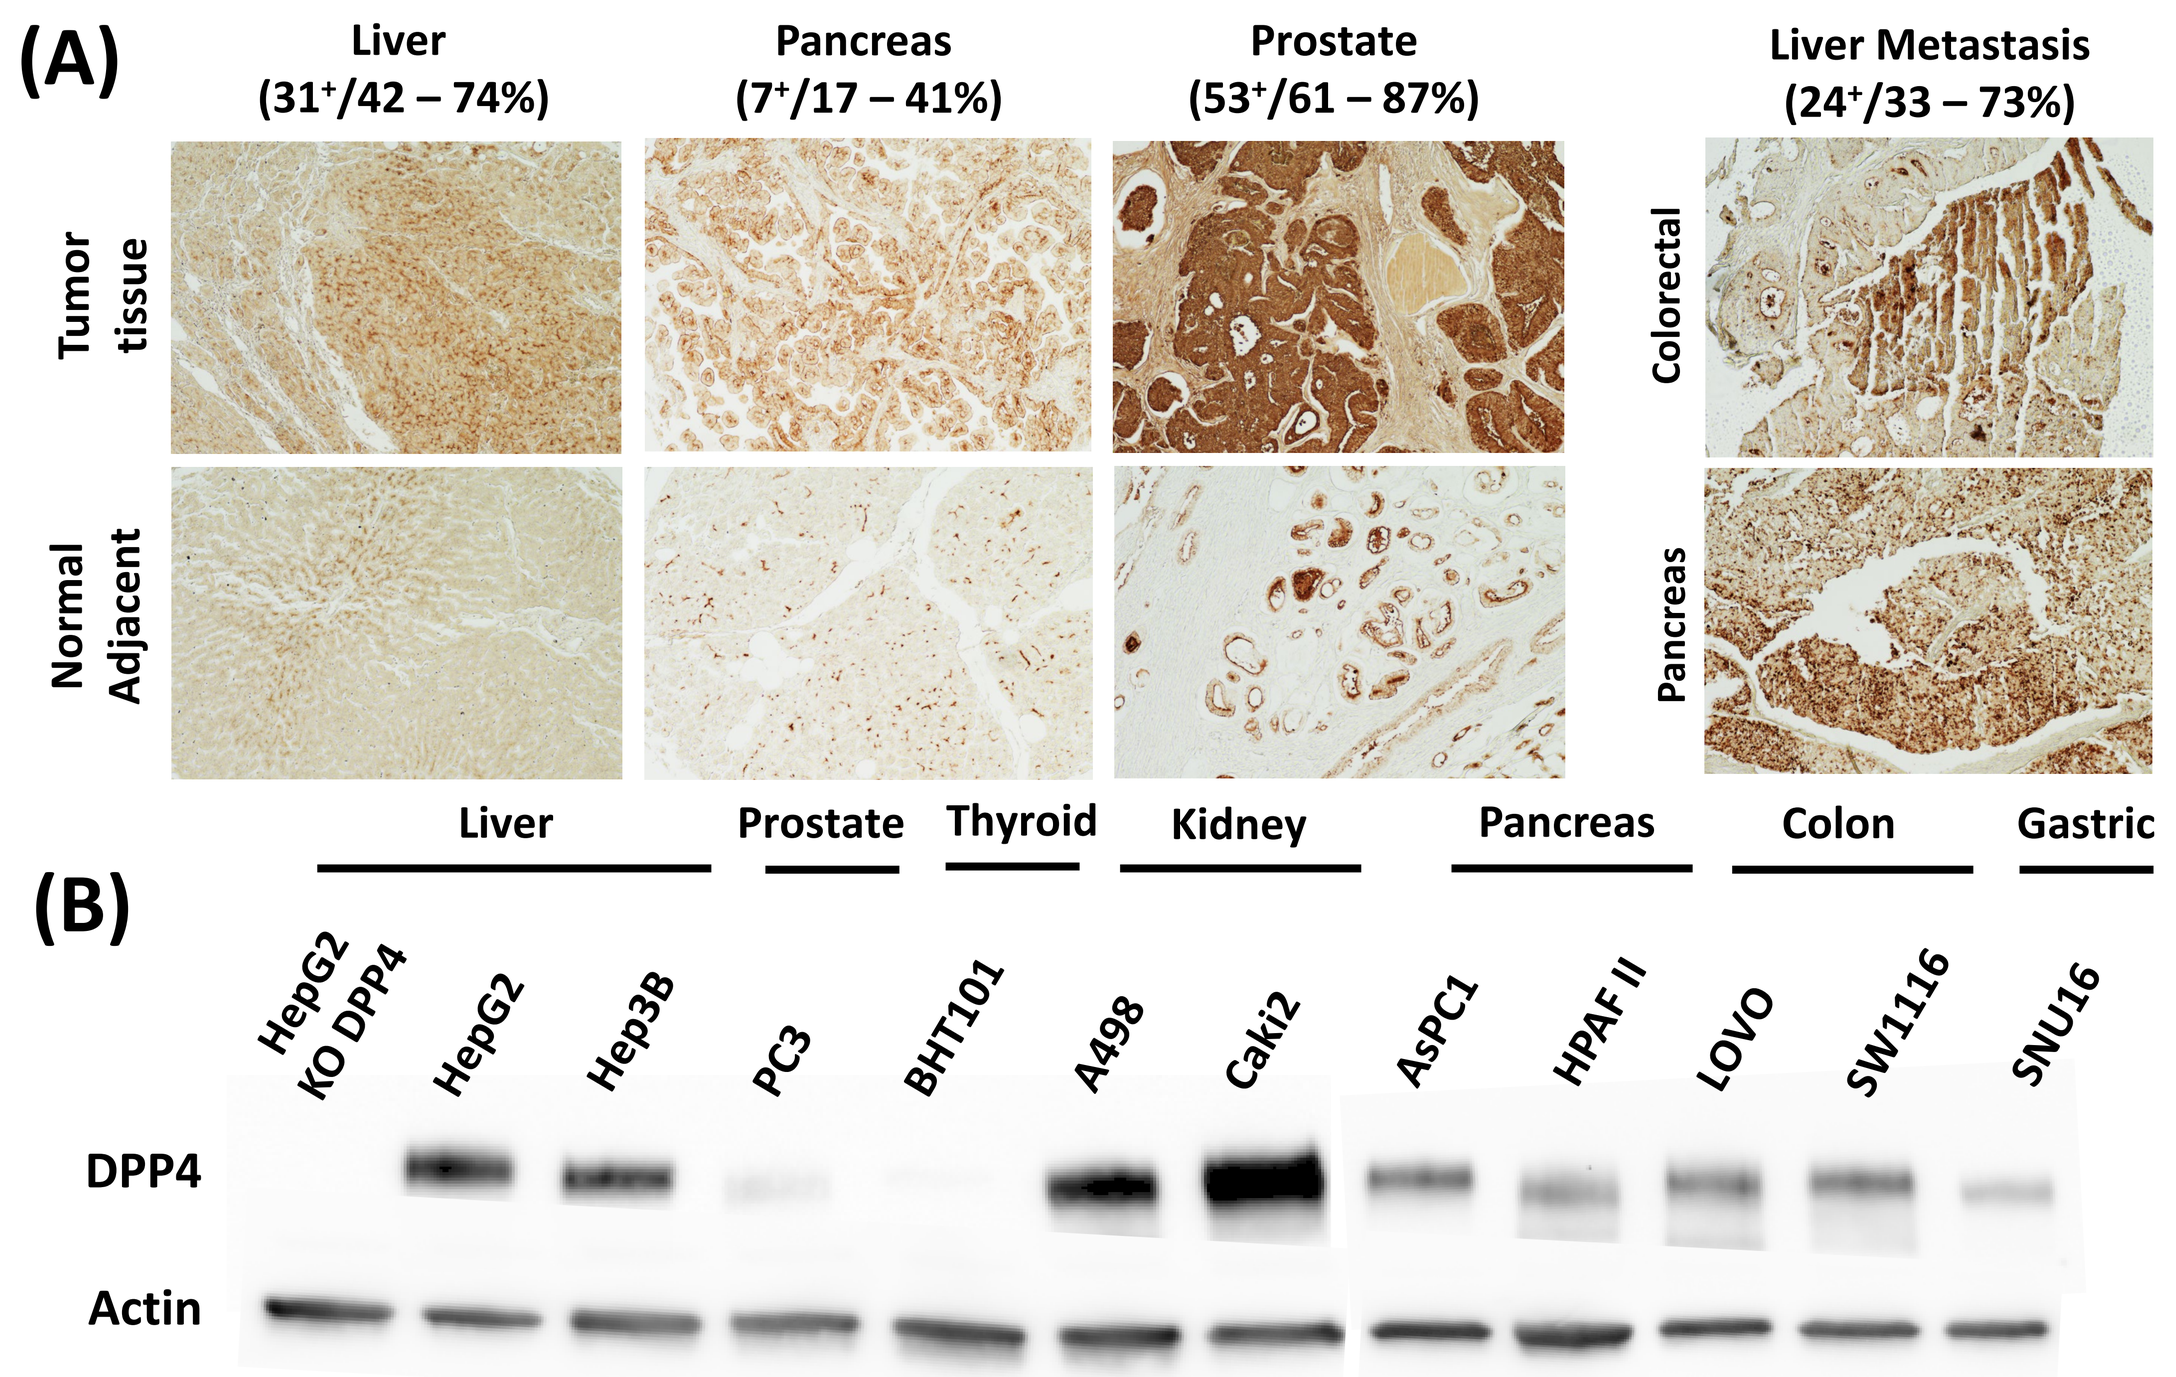

(A)
Liver (31+/42 – 74%)
Pancreas (7+/17 – 41%)
Prostate (53+/61 – 87%)
Liver Metastasis (24+/33 – 73%)
Tumor tissue
Normal Adjacent
Colorectal
Pancreas
(B)
Liver
Prostate
Thyroid
Kidney
Pancreas
Colon
Gastric
HepG2 KO DPP4
HepG2
Hep3B
PC3
BHT101
A498
Caki2
AsPC1
HPAF II
LOVO
SW1116
SNU16
DPP4
Actin

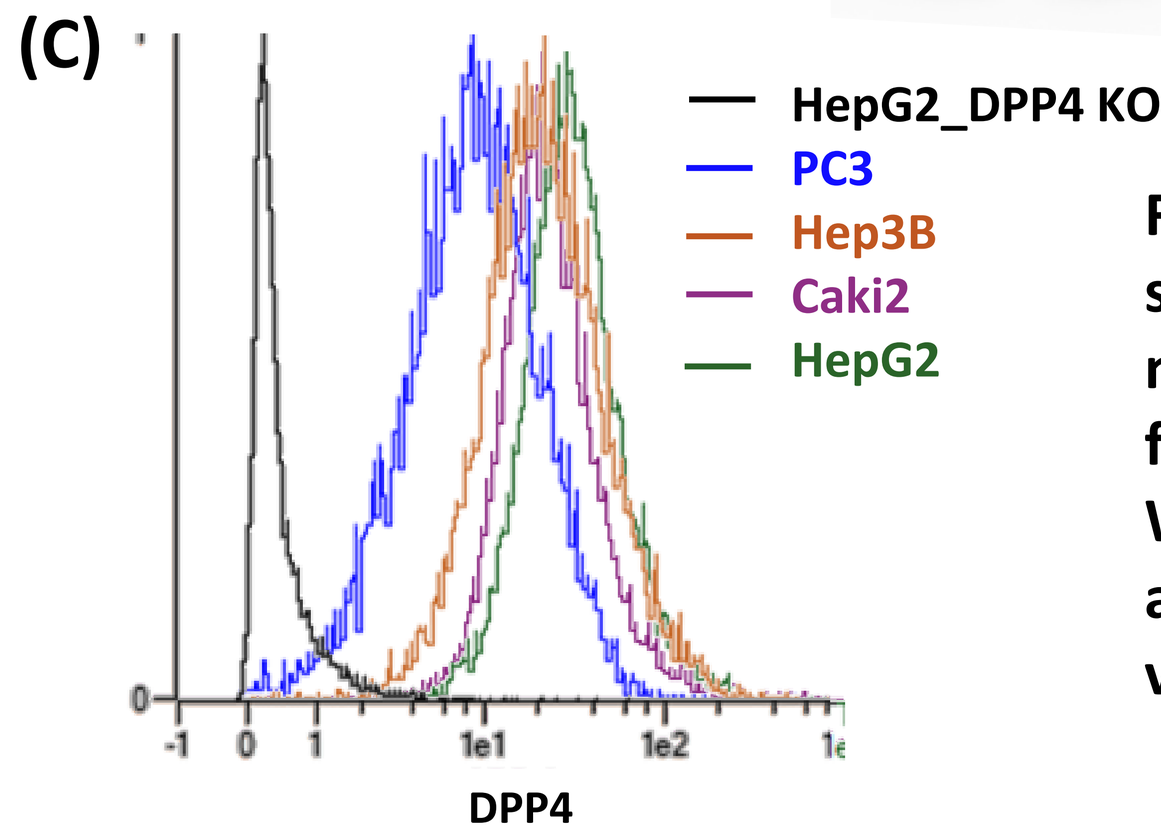

(C)
HepG2_DPP4 KO
PC3
Hep3B
Caki2
HepG2
DPP4

Fig.2 (A) DPP4 expression by IHC staining (commercial anti-DPP4 mAb) on TMA of human tumors and frequency of DPP4+ samples. (B) Western blot and (C) flow cytometry analyses of DPP4 expression on various human tumor cell lines.

3. DPP4 is overexpressed in senescent cancer cells

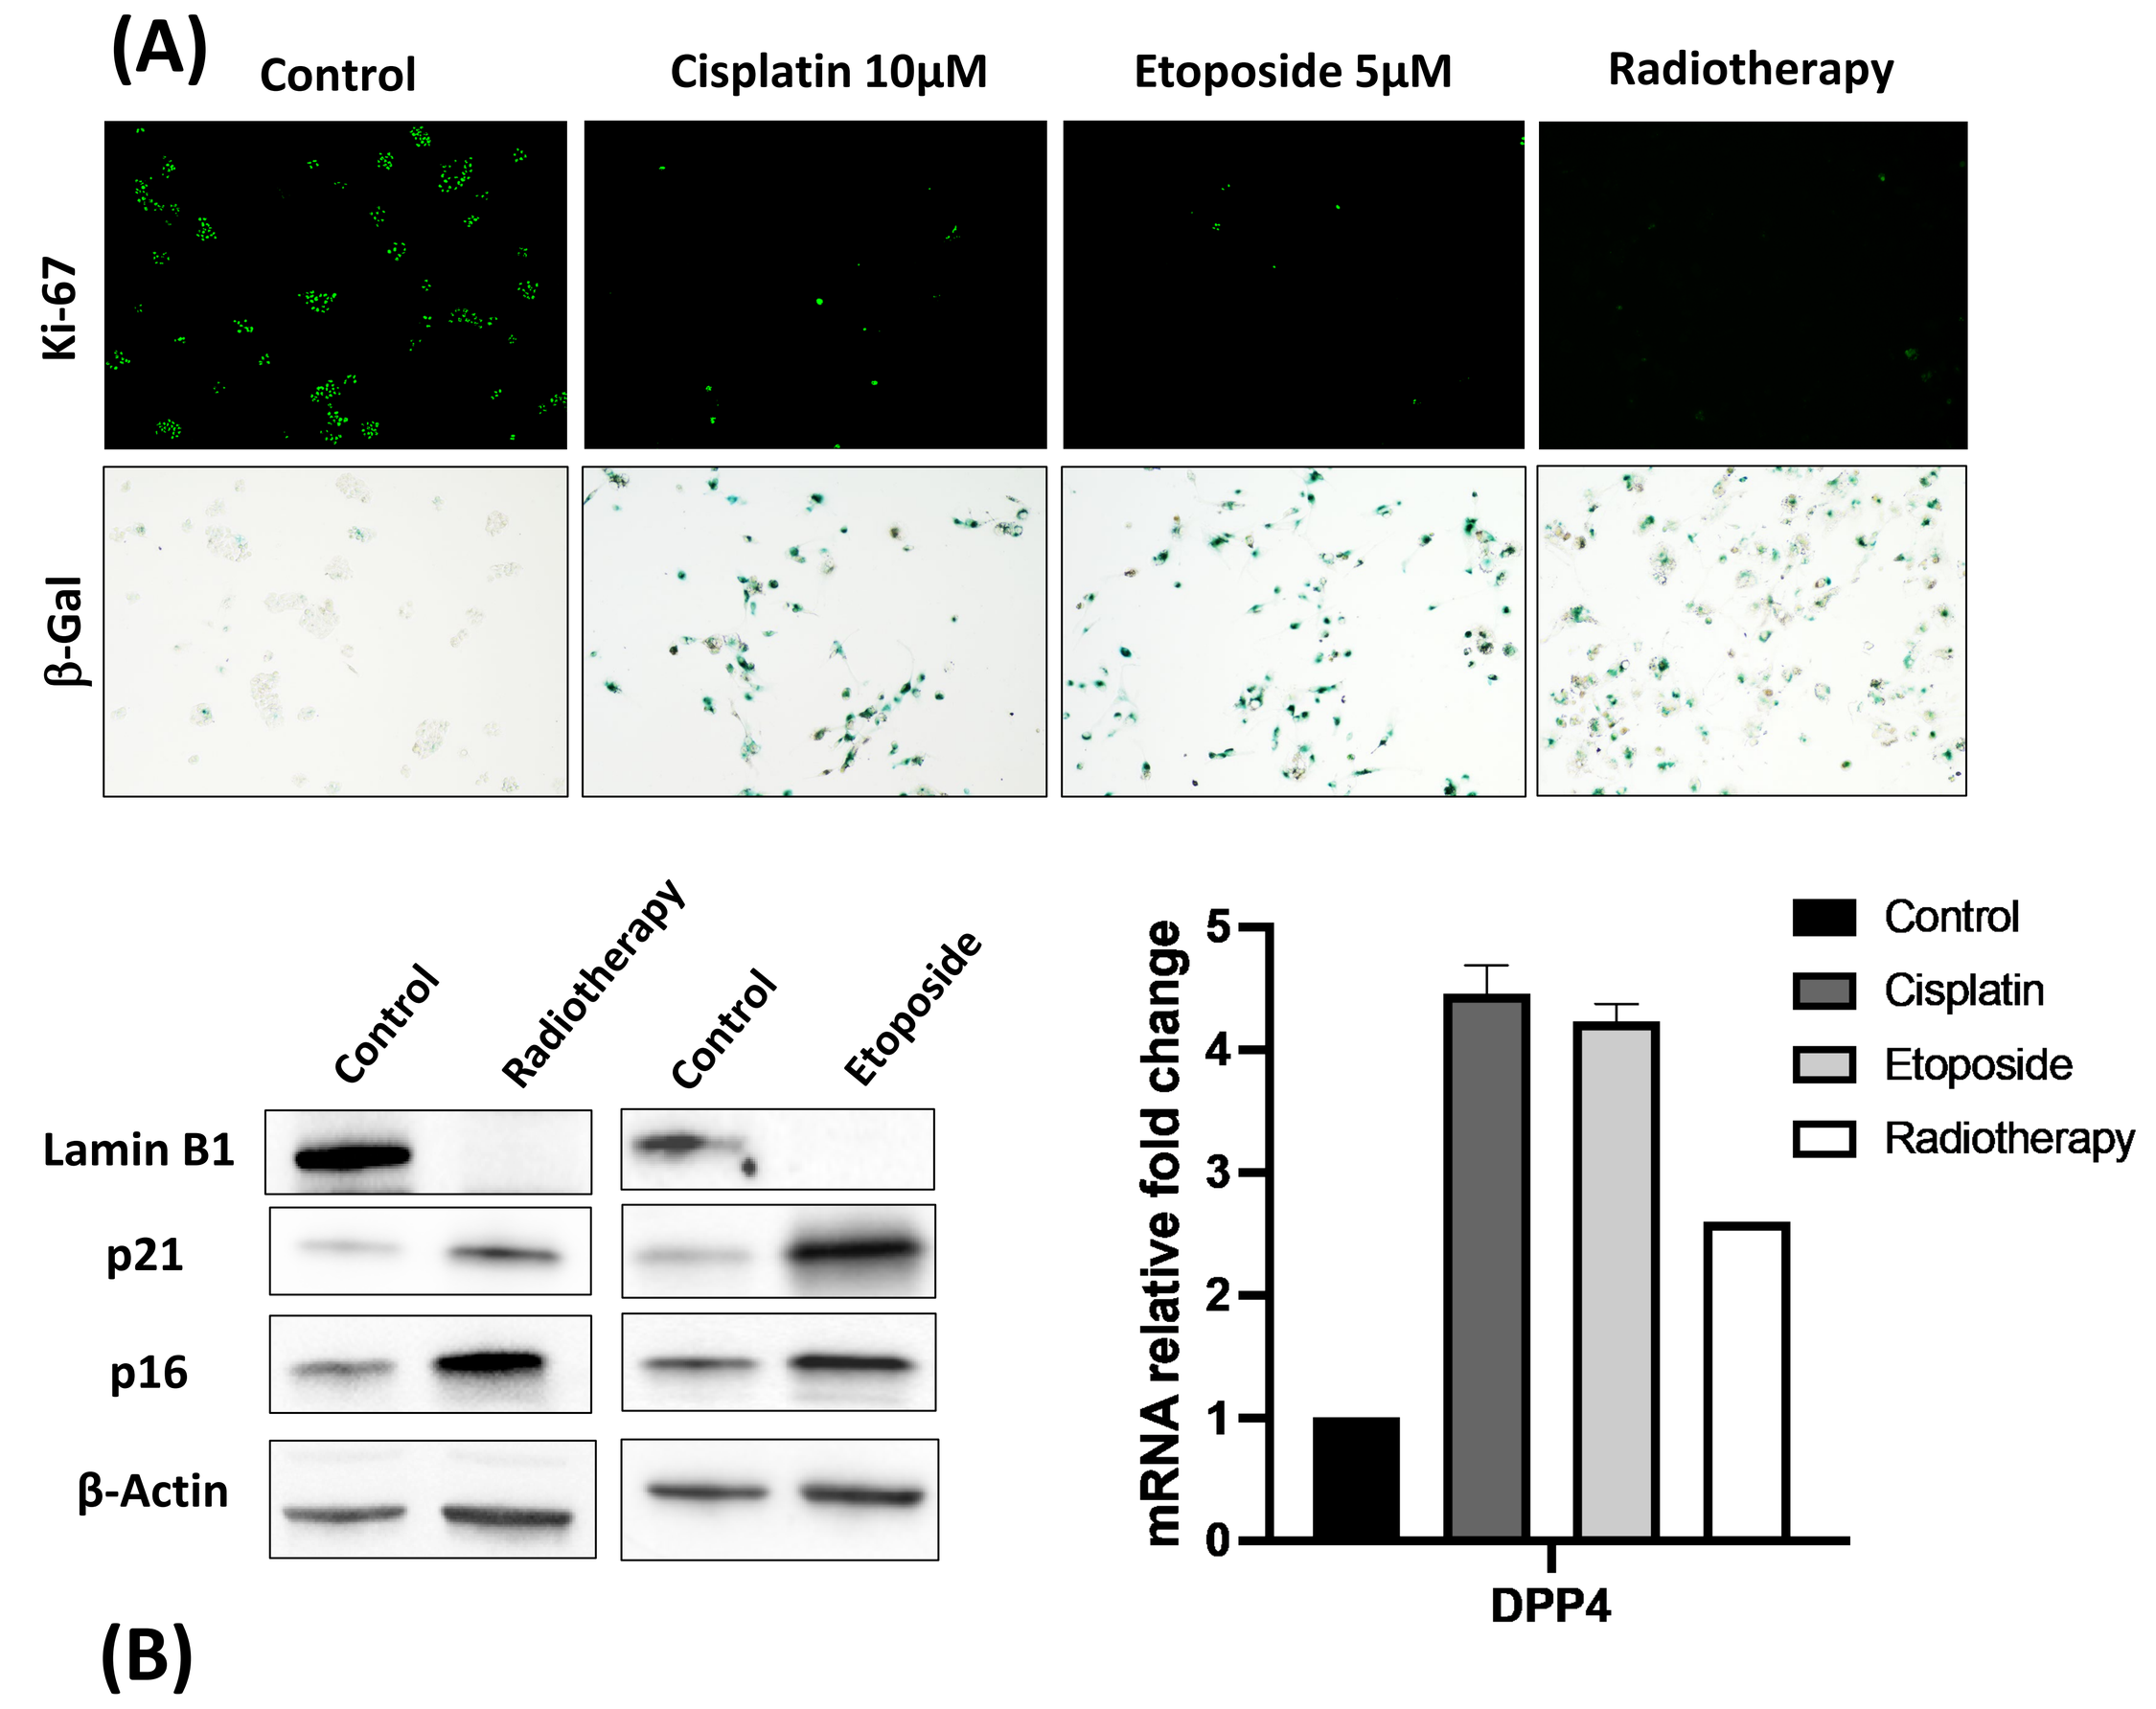

(A)
Control
Cisplatin 10µM
Etoposide 5µM
Radiotherapy
Ki-67
β-Gal
Control
Radiotherapy
Control
Etoposide
Lamin B1
p21
p16
β-Actin
mRNA relative fold change
DPP4
Control
Cisplatin
Etoposide
Radiotherapy

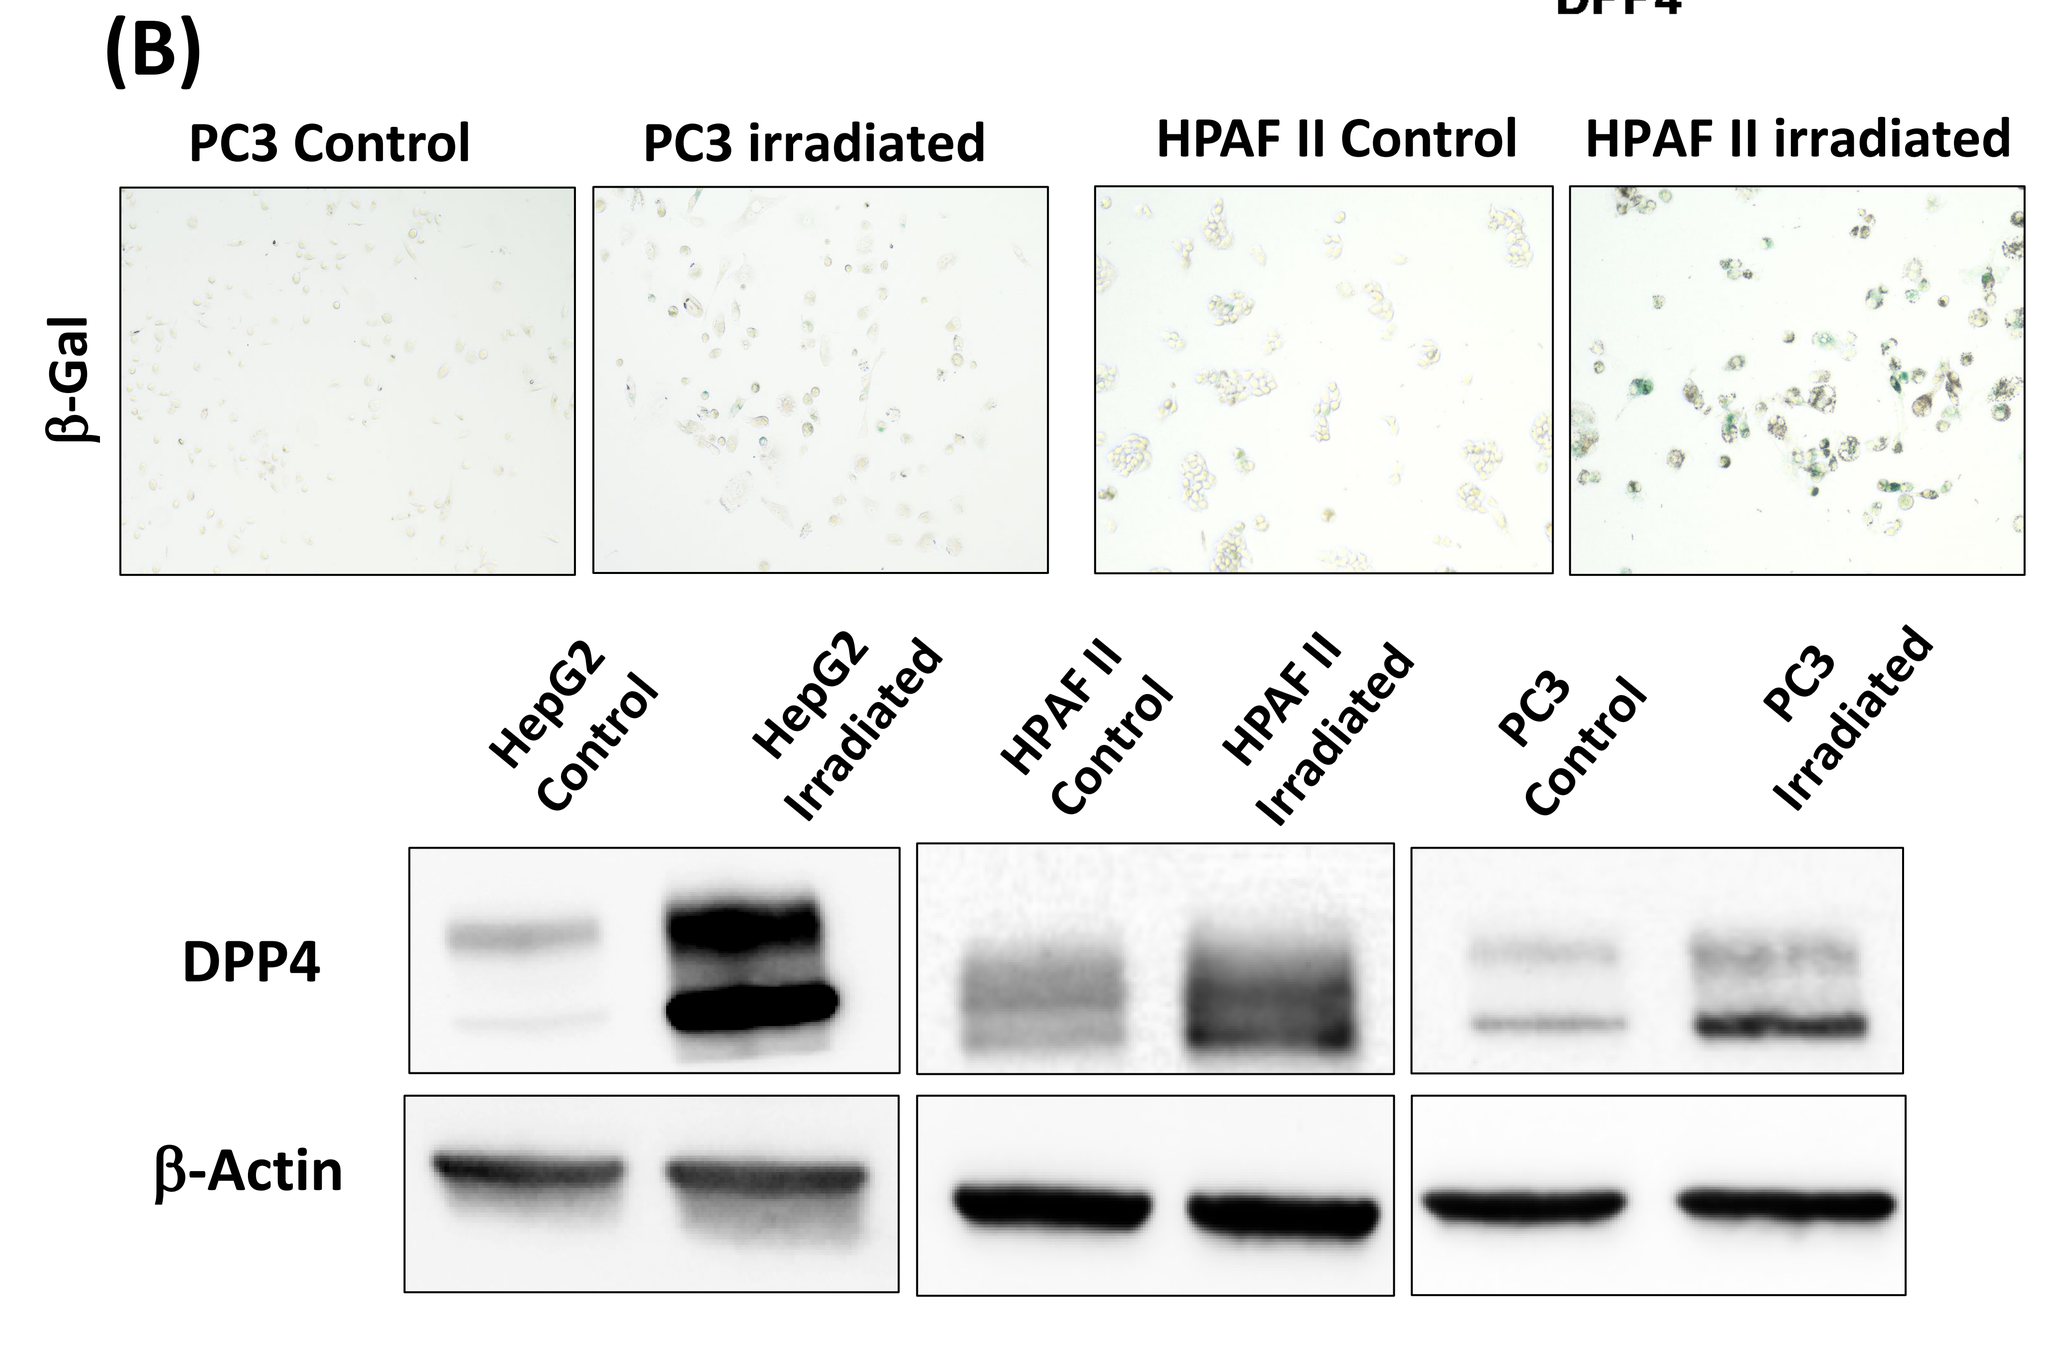

(B)
PC3 Control
PC3 irradiated
HPAF II Control
HPAF II irradiated
β-Gal
HepG2 Control
HepG2 Irradiated
HPAF II Control
HPAF II Irradiated
PC3 Control
PC3 Irradiated
DPP4
β-Actin

Fig.3 (A) Senescence induction of human tumor cell lines by radiotherapy/chemotherapies (HepG2) or (B) radiatherapy (HepG2, HPAF II, PC3) leading to overexpression of DPP4.

4. High-affinity binding of STX-1 anti-DPP4 mAb

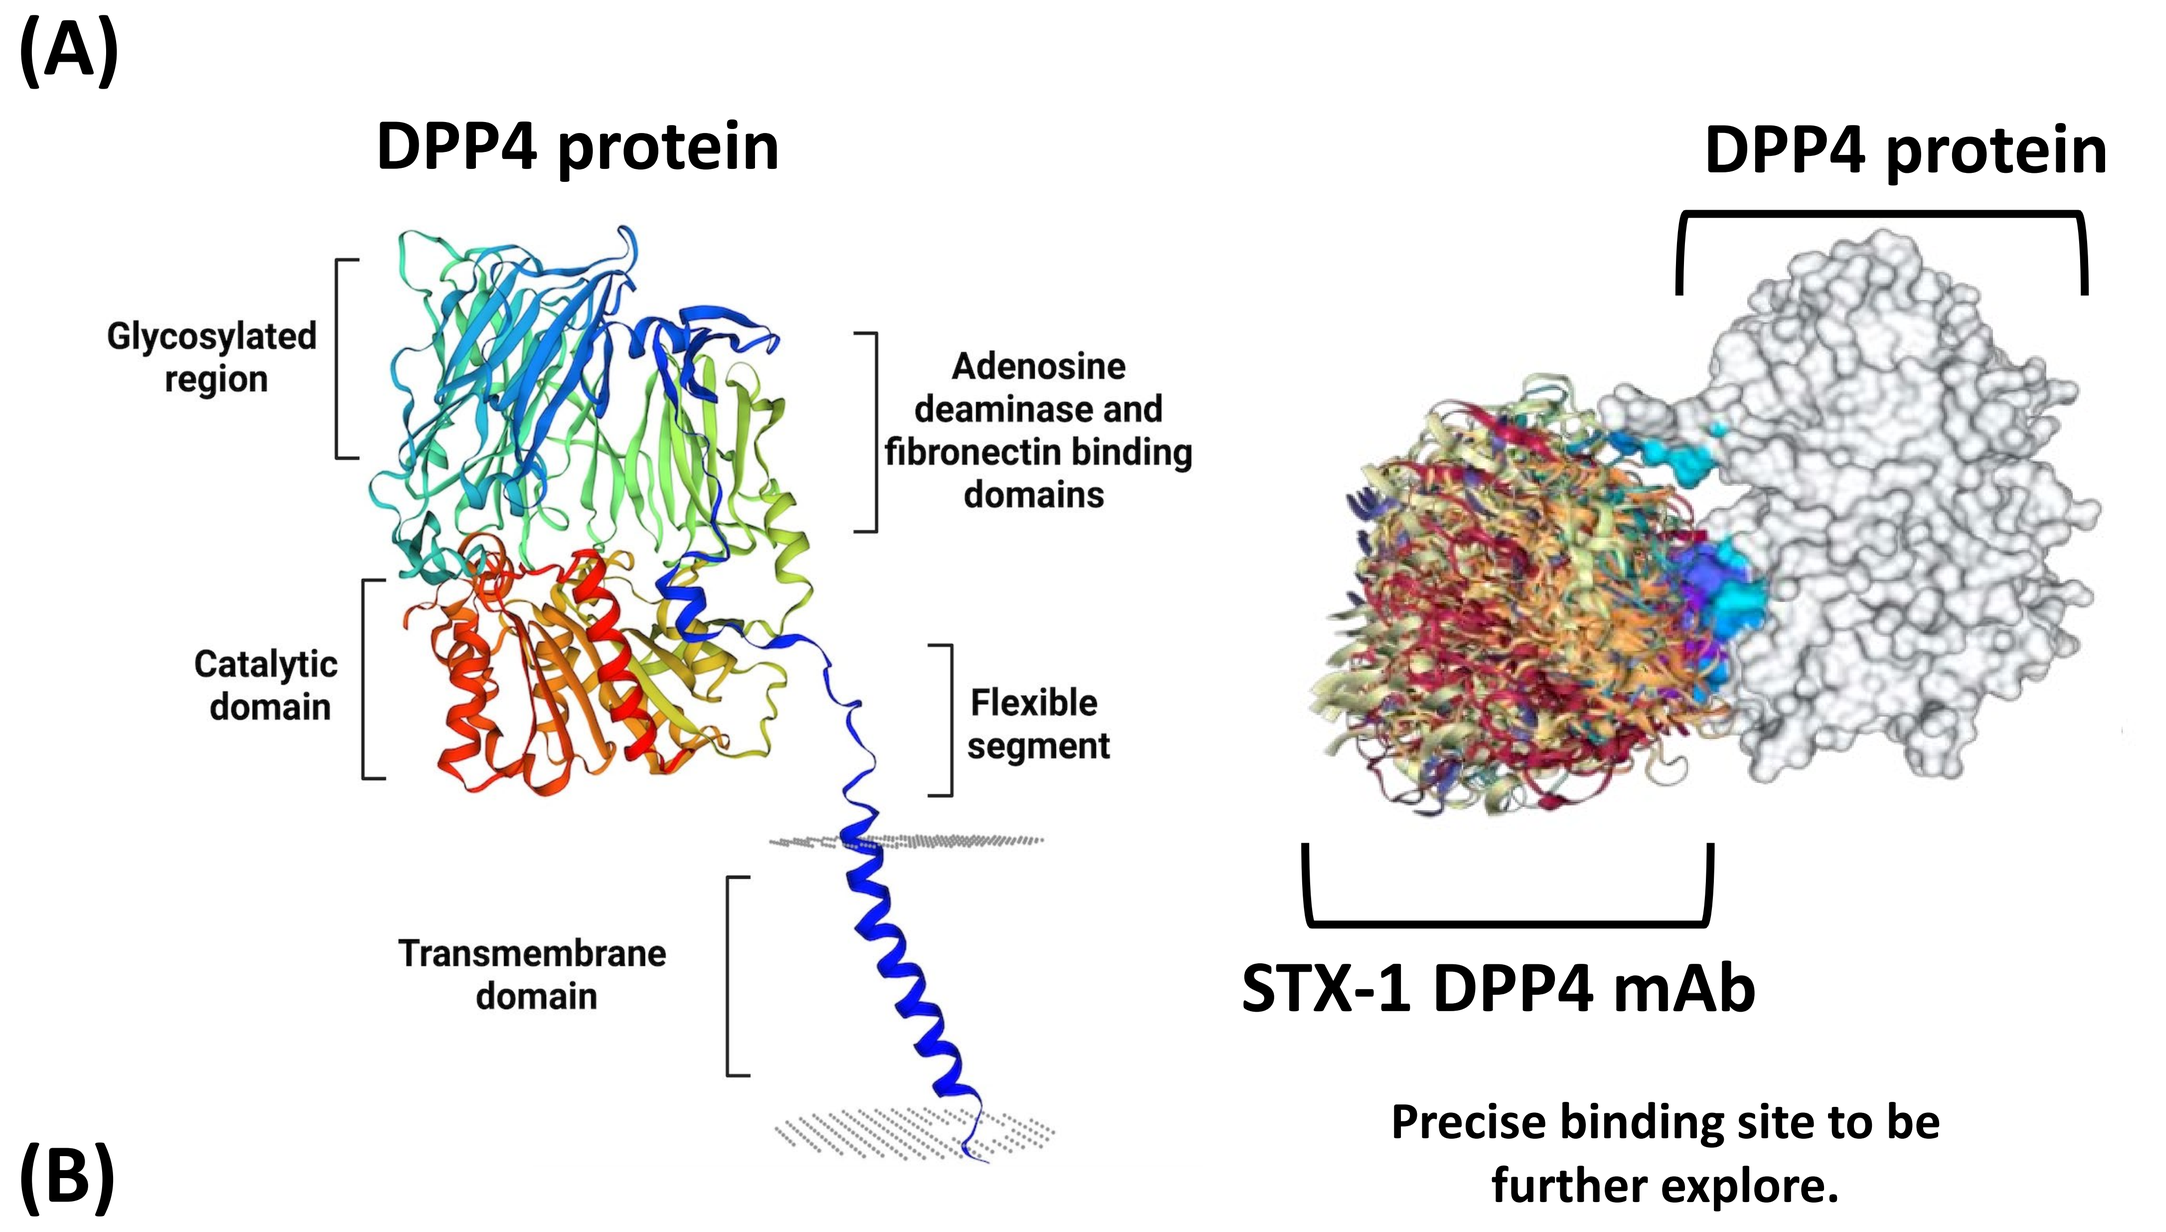

(A)
DPP4 protein
Glycosylated region
Adenosine deaminase and fibronectin binding domains
Catalytic domain
Flexible segment
Transmembrane domain
DPP4 protein
STX-1 DPP4 mAb
Precise binding site to be further explore.

(B)

	K_{on} ($M^{-1}.s^{-1}$)	K_{off} (s^{-1})	K_D (M)
DPP4 antibody	$4{,}3.10^{4}$	$6{,}3.10^{-9}$	$1{,}46.10^{-13}$

Fig.4 (A) *In silico* epitope mapping between STX-1 mAb and the human DPP4 membrane protein and (B) affinity constants of the STX-1 mAb (association rate (K_{on}), dissociation rate (K_{off}) and K_D equilibrium constant (ratio K_{off}/K_{on}) by using Biacore assay.

5. STX-1 free mAb and ADCs are efficiently internalized

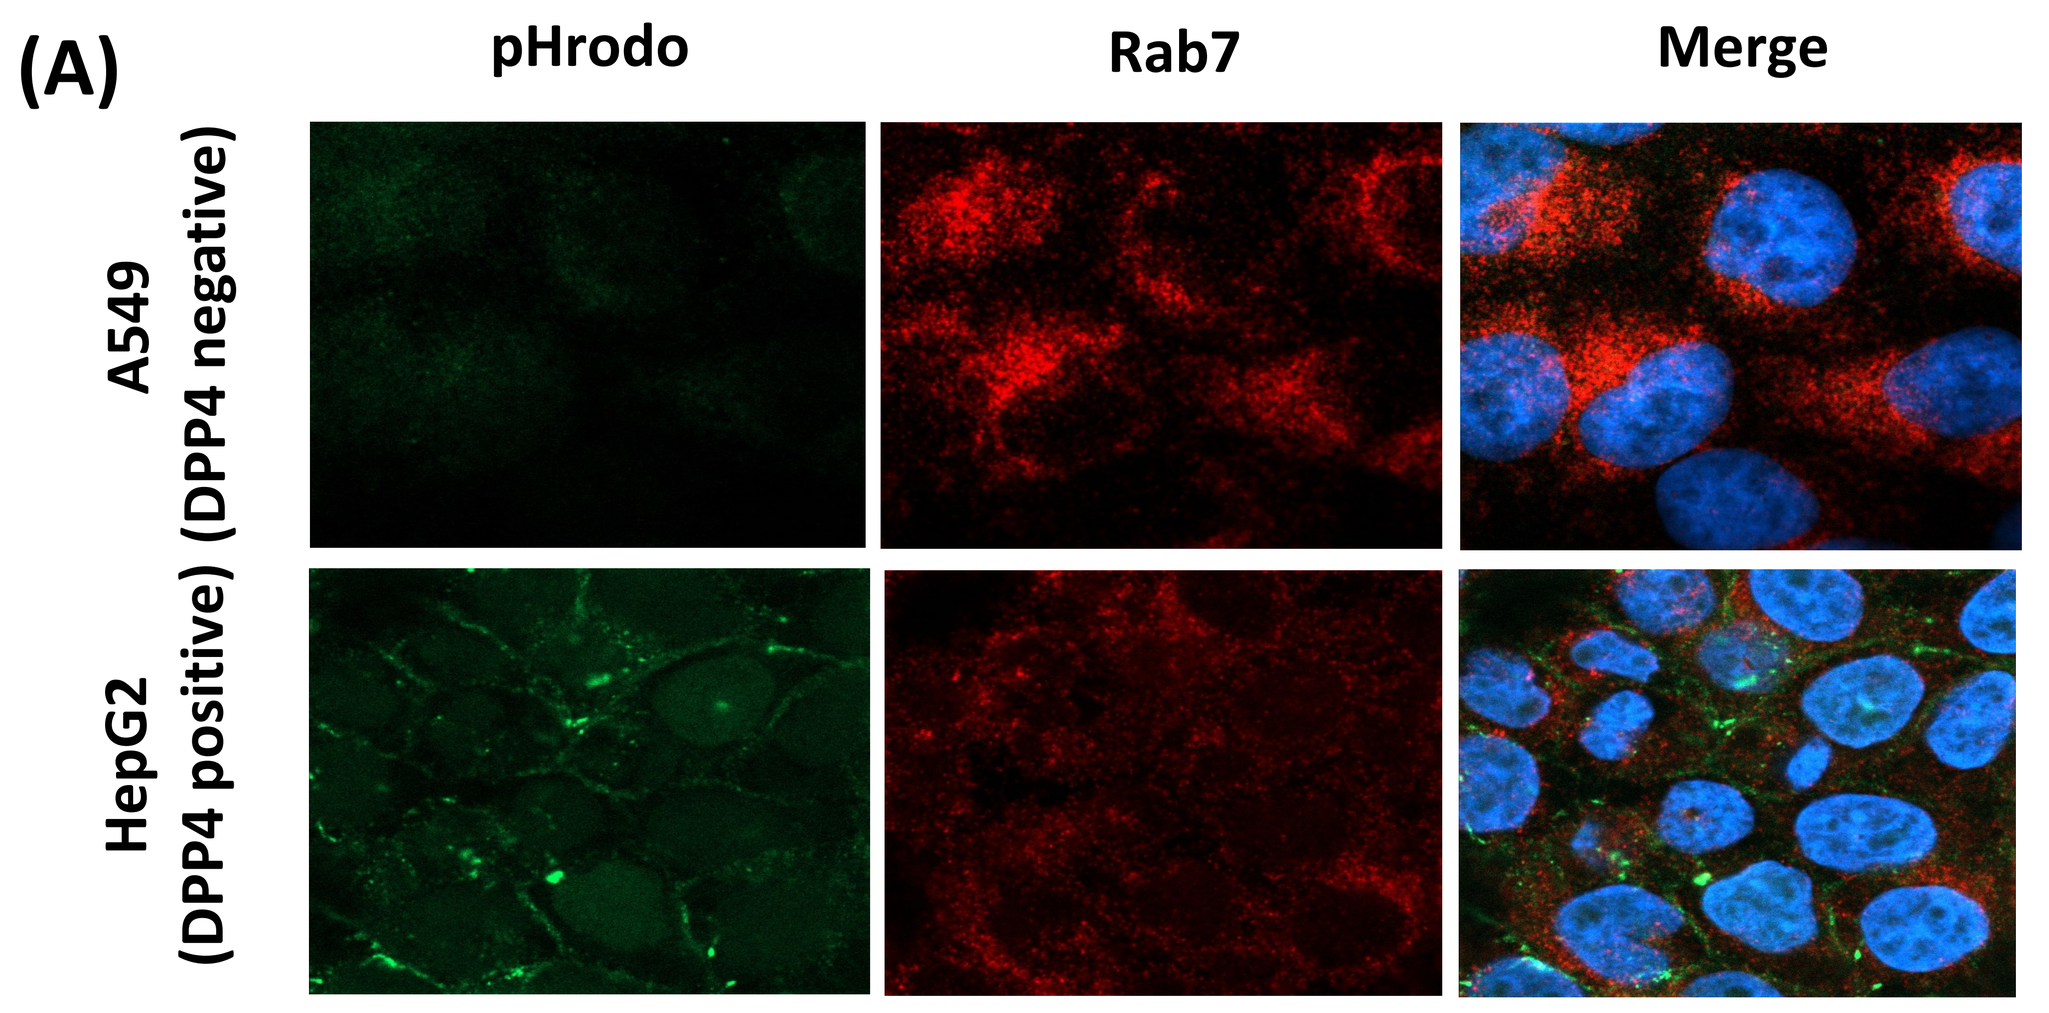

(A)
pHrodo
Rab7
Merge
A549 (DPP4 negative)
HepG2 (DPP4 positive)

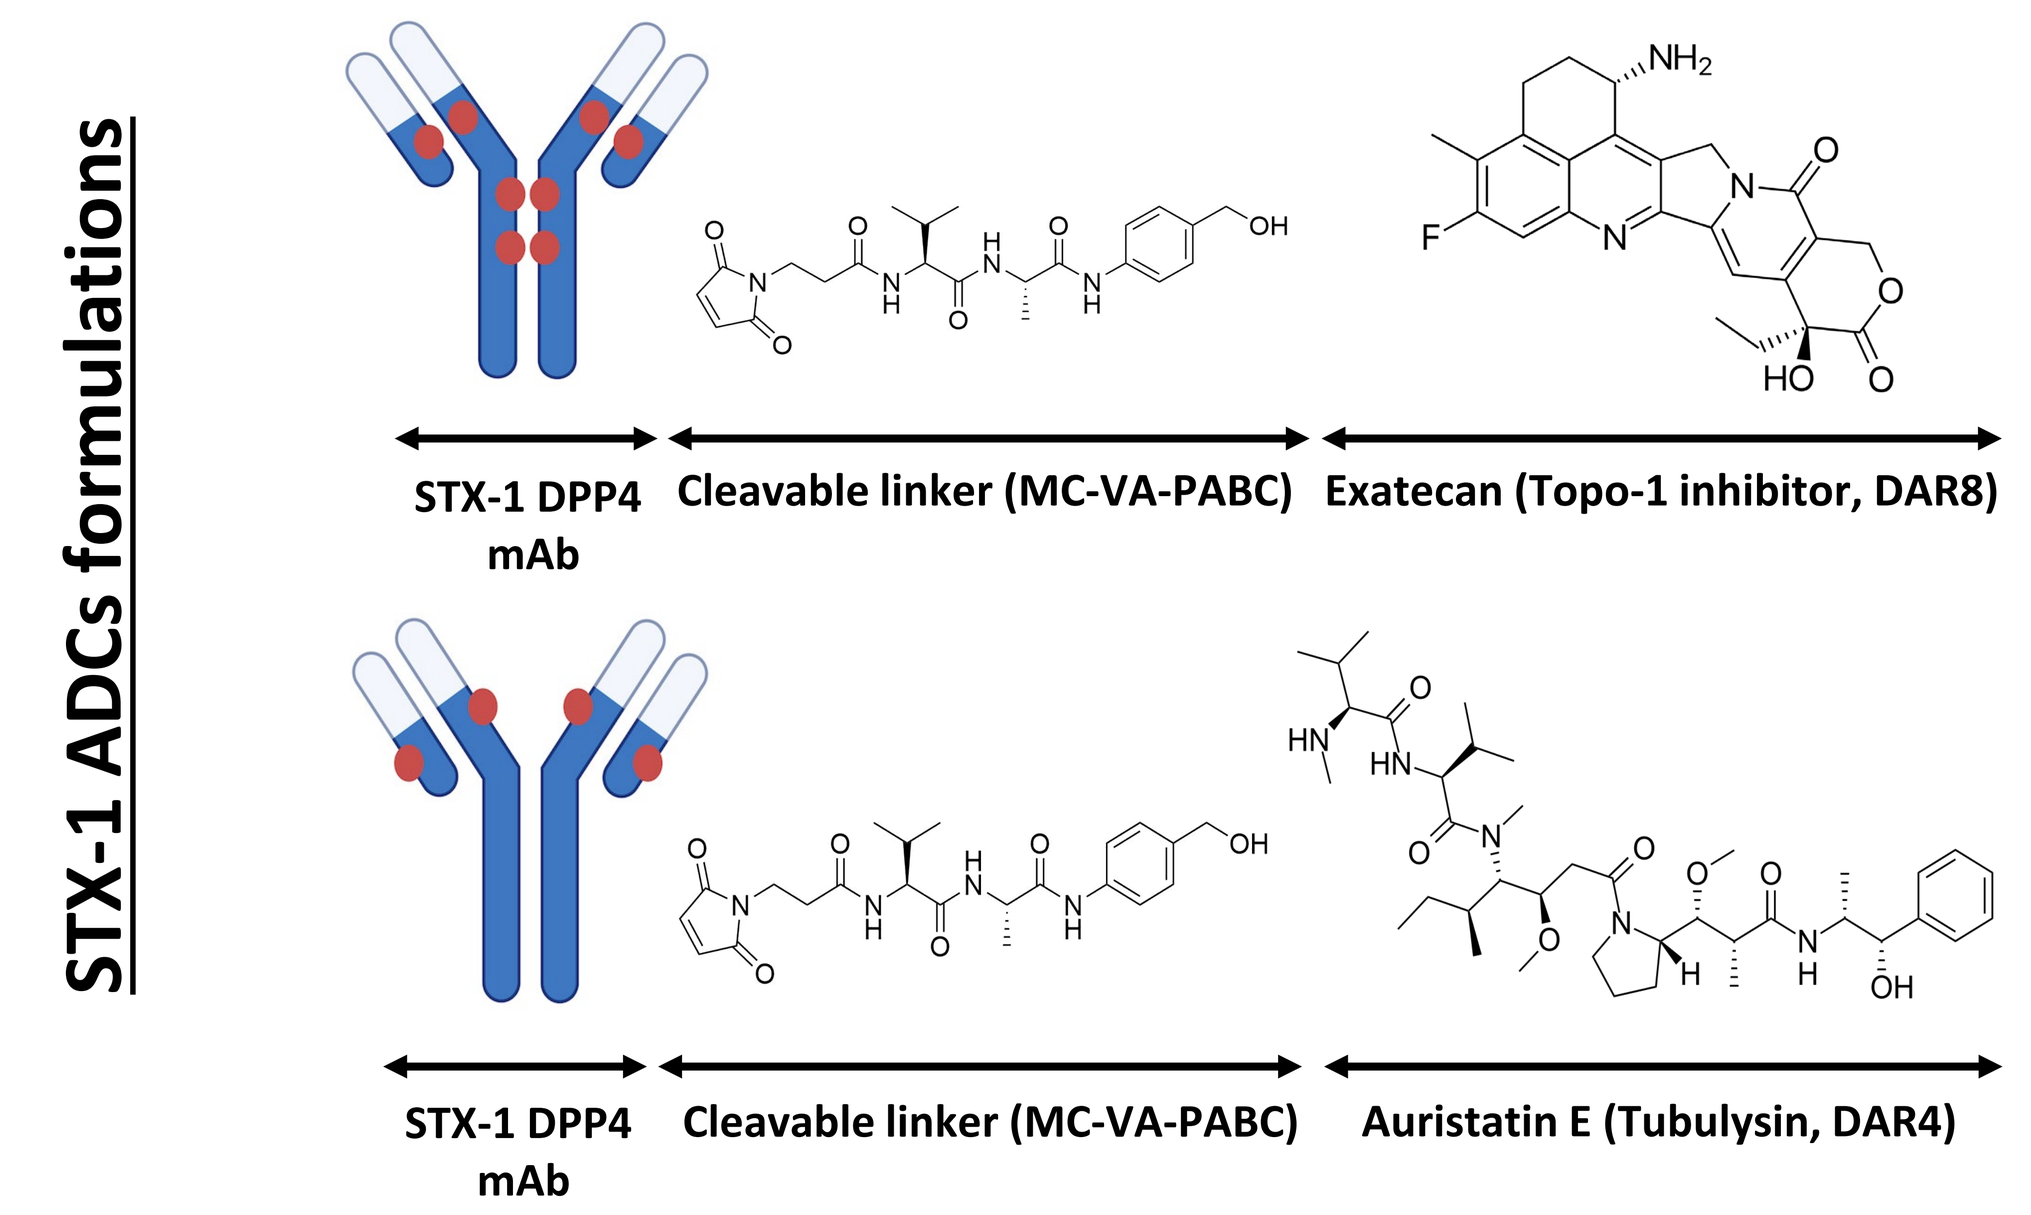

STX-1 ADCs formulations
STX-1 DPP4 mAb
Cleavable linker (MC-VA-PABC)
Exatecan (Topo-1 inhibitor, DAR8)
STX-1 DPP4 mAb
Cleavable linker (MC-VA-PABC)
Auristatin E (Tubulysin, DAR4)

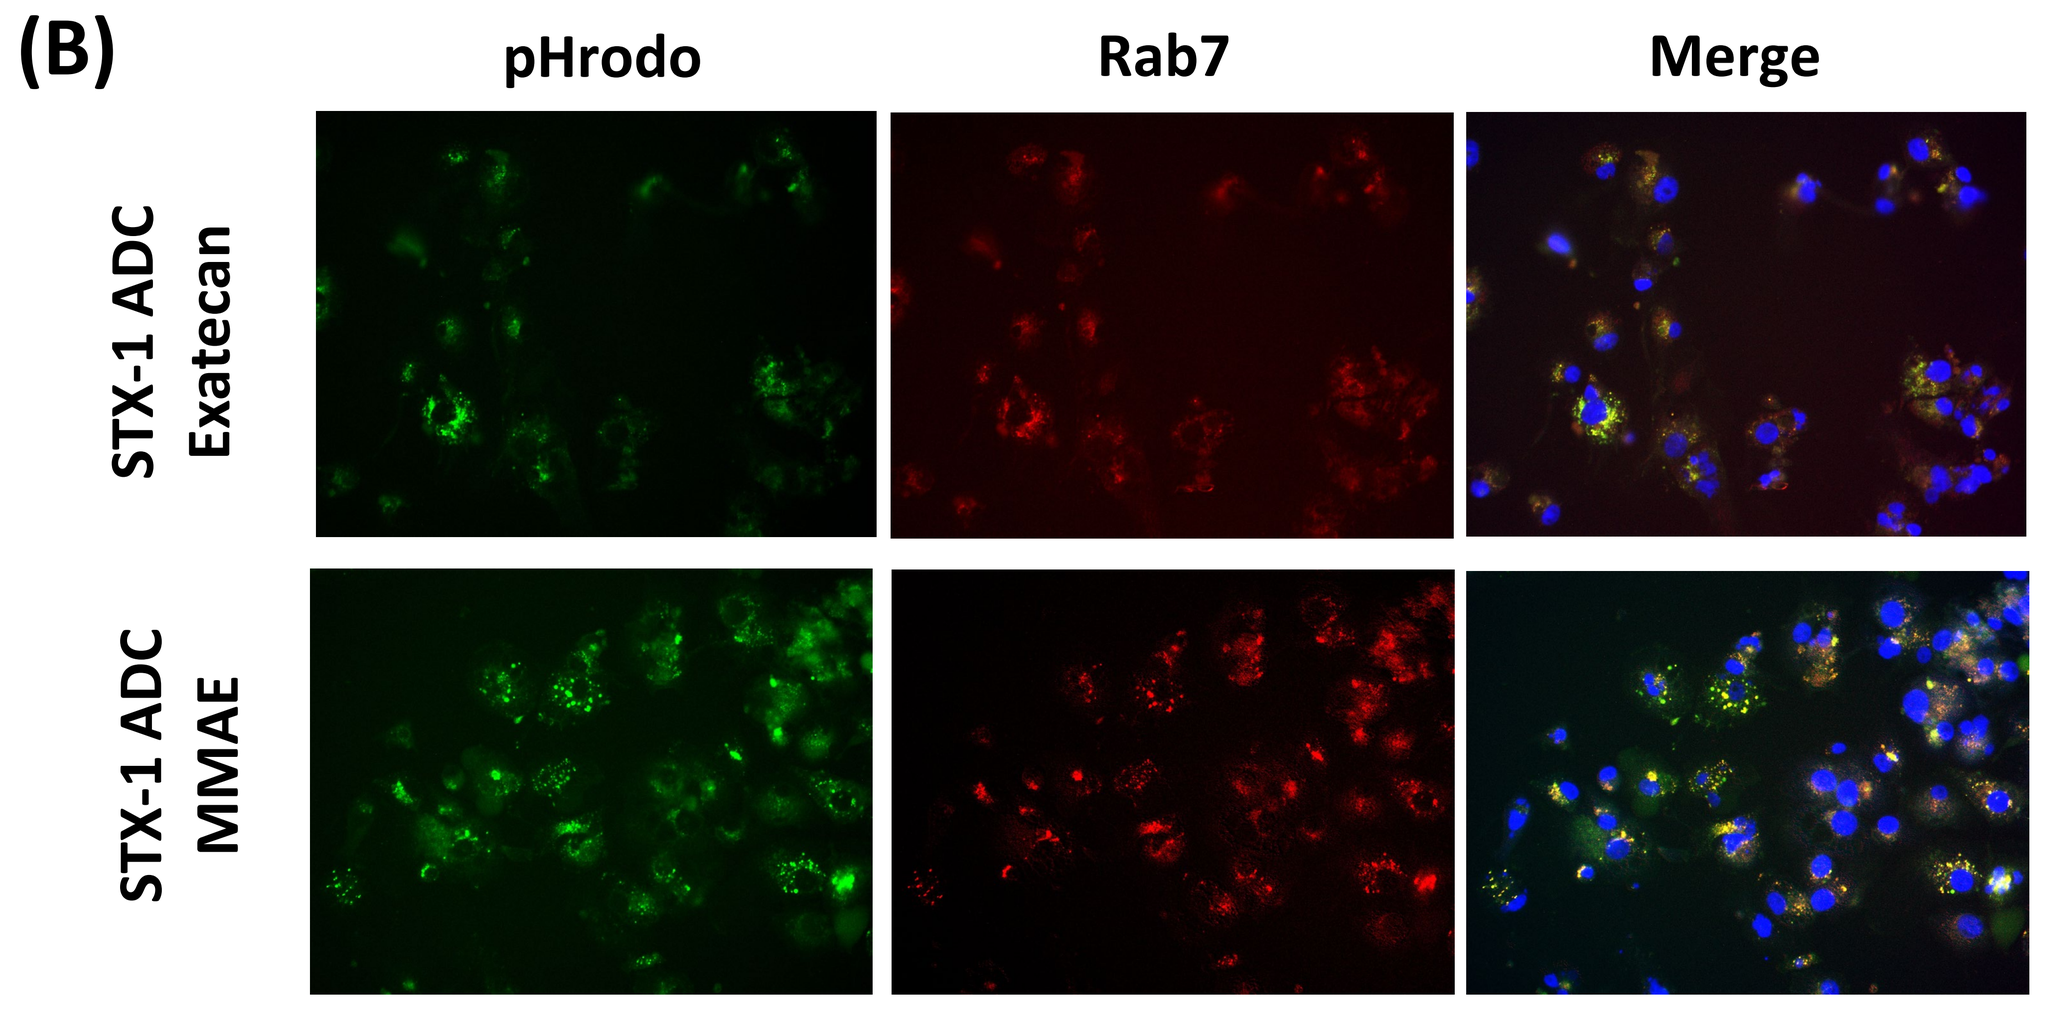

(B)
pHrodo
Rab7
Merge
STX-1 ADC Exatecan
STX-1 ADC MMAE

Fig.5 (A) Internalization of free DPP4 mAb in HepG2 and A549 cancer cell lines after 24h. (B) Internalization of STX-1 ADCs after 40h in HepG2 senescent cells induced by 20 Gy radiation.

6. STX-1 ADCs are effective *in vitro* on cancer cells

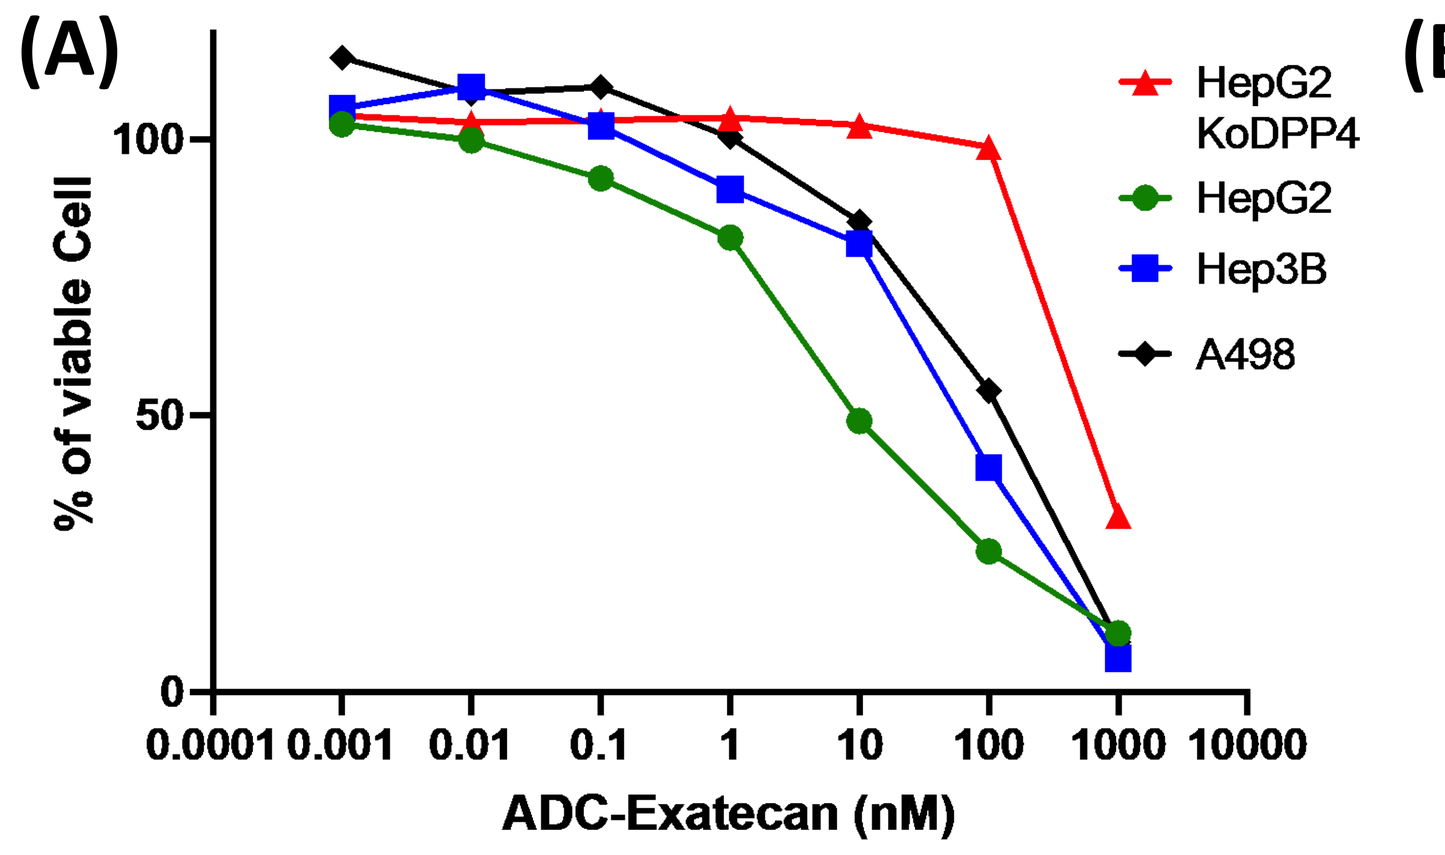

(A)
% of viable Cell
ADC-Exatecan (nM)
HepG2 KoDPP4
HepG2
Hep3B
A498

(B)

Cell lines	IC50 (nM)
HepG2	6,4
Hep3B	15,65
A498	109
HepG2 KO DPP4	449

Fig.6 (A) *In vitro* cell viability and (B) IC50 in nM after ADC–Exatecan treatment on DPP4 positive/negative cancer cell lines.

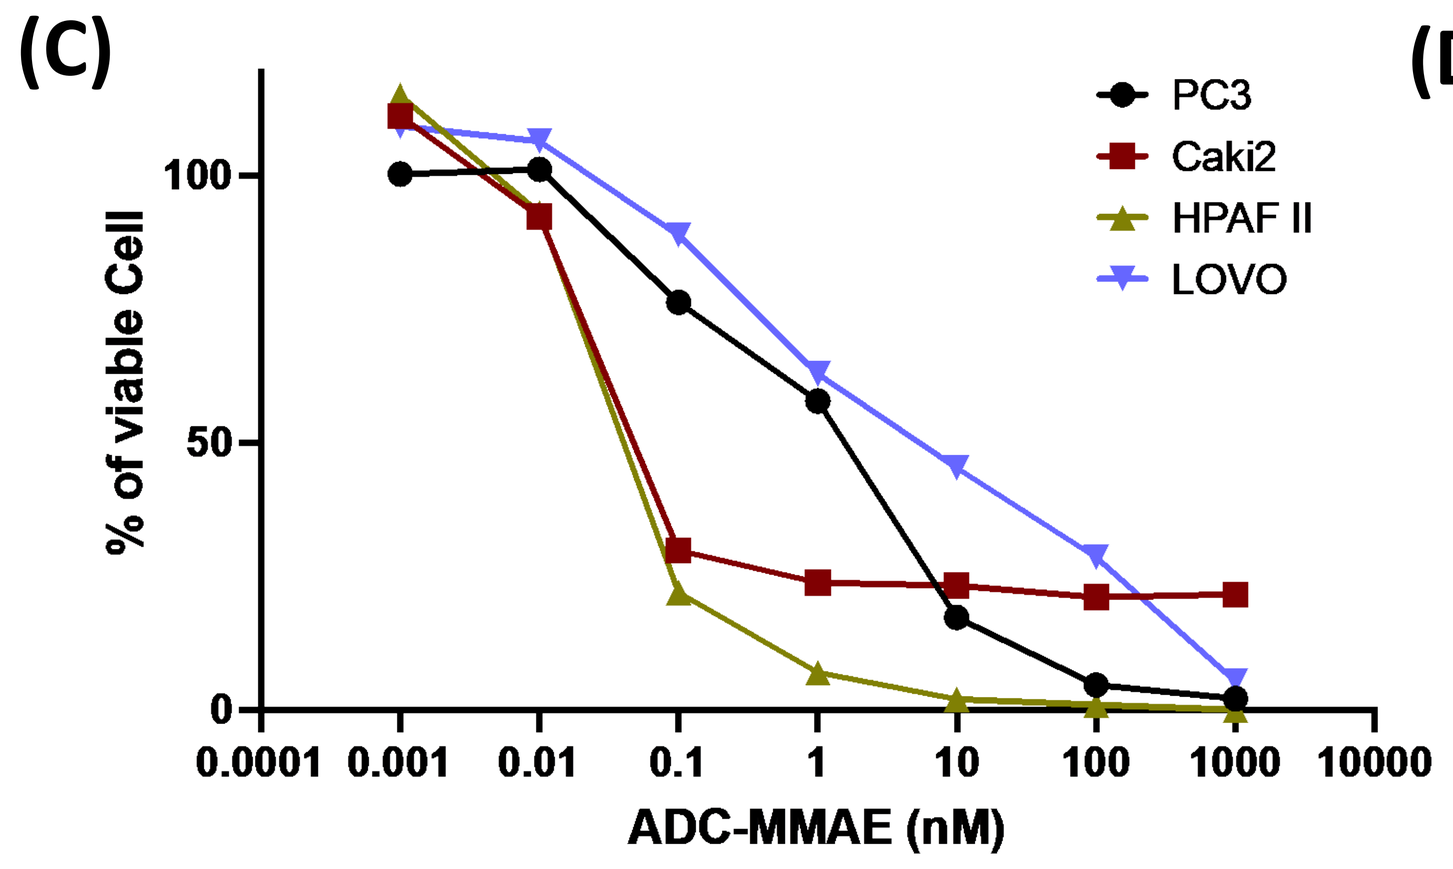

(C)
% of viable Cell
ADC-MMAE (nM)
PC3
Caki2
HPAF II
LOVO

(D)

Cell lines	IC50 (nM)
PC3	1,27
Caki2	0,063
HPAF II	0,046
LOVO	1,17

Fig.6 (C) *In vitro* cell viability and (D) IC50 in nM after ADC–MMAE treatment on DPP4 positive cancer cell lines.

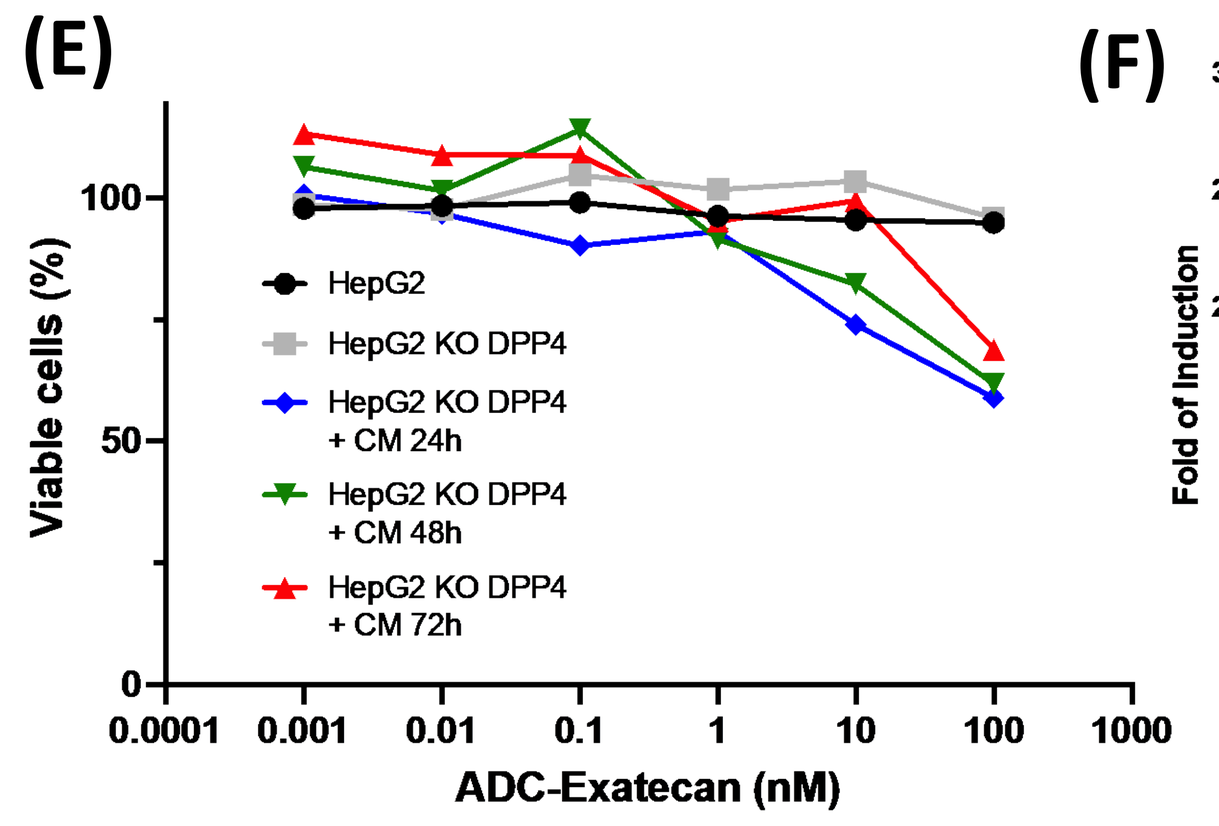

(E)
Viable cells (%)
ADC-Exatecan (nM)
HepG2
HepG2 KO DPP4
HepG2 KO DPP4 + CM 24h
HepG2 KO DPP4 + CM 48h
HepG2 KO DPP4 + CM 72h

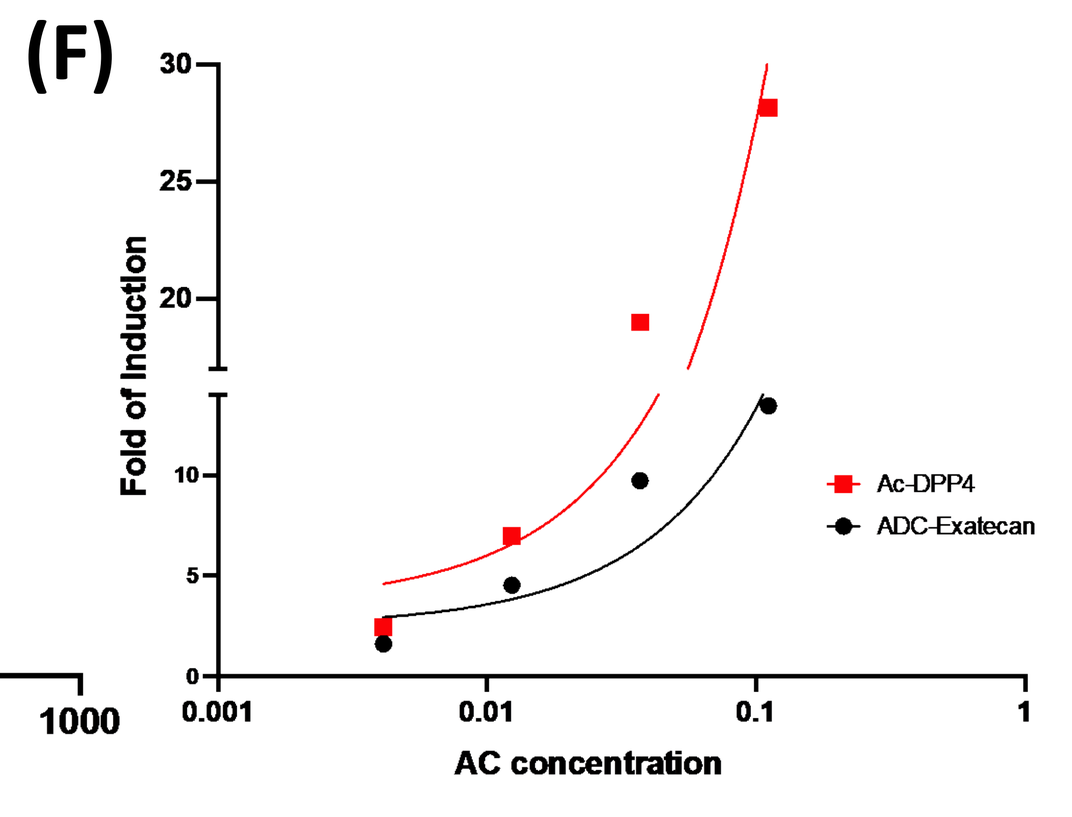

(F)
Fold of Induction
AC concentration
Ac-DPP4
ADC-Exatecan

Fig.6 (E) Bystander killing effect on HepG2 Ko-DPP4 viability following treatment with CM from HepG2 treated cells. CM were generated by exposing HepG2 cells to ADC-Exatecan for 24 hours, 48 hours, or 72 hours. (F) ADCC activity with ADC-Exatecan and AC-DPP4 on HepG2/Jurkat-FcγRIIIa cells co-culture (ratio 1/2).

7. STX-1 ADCs reduced significantly tumor growth

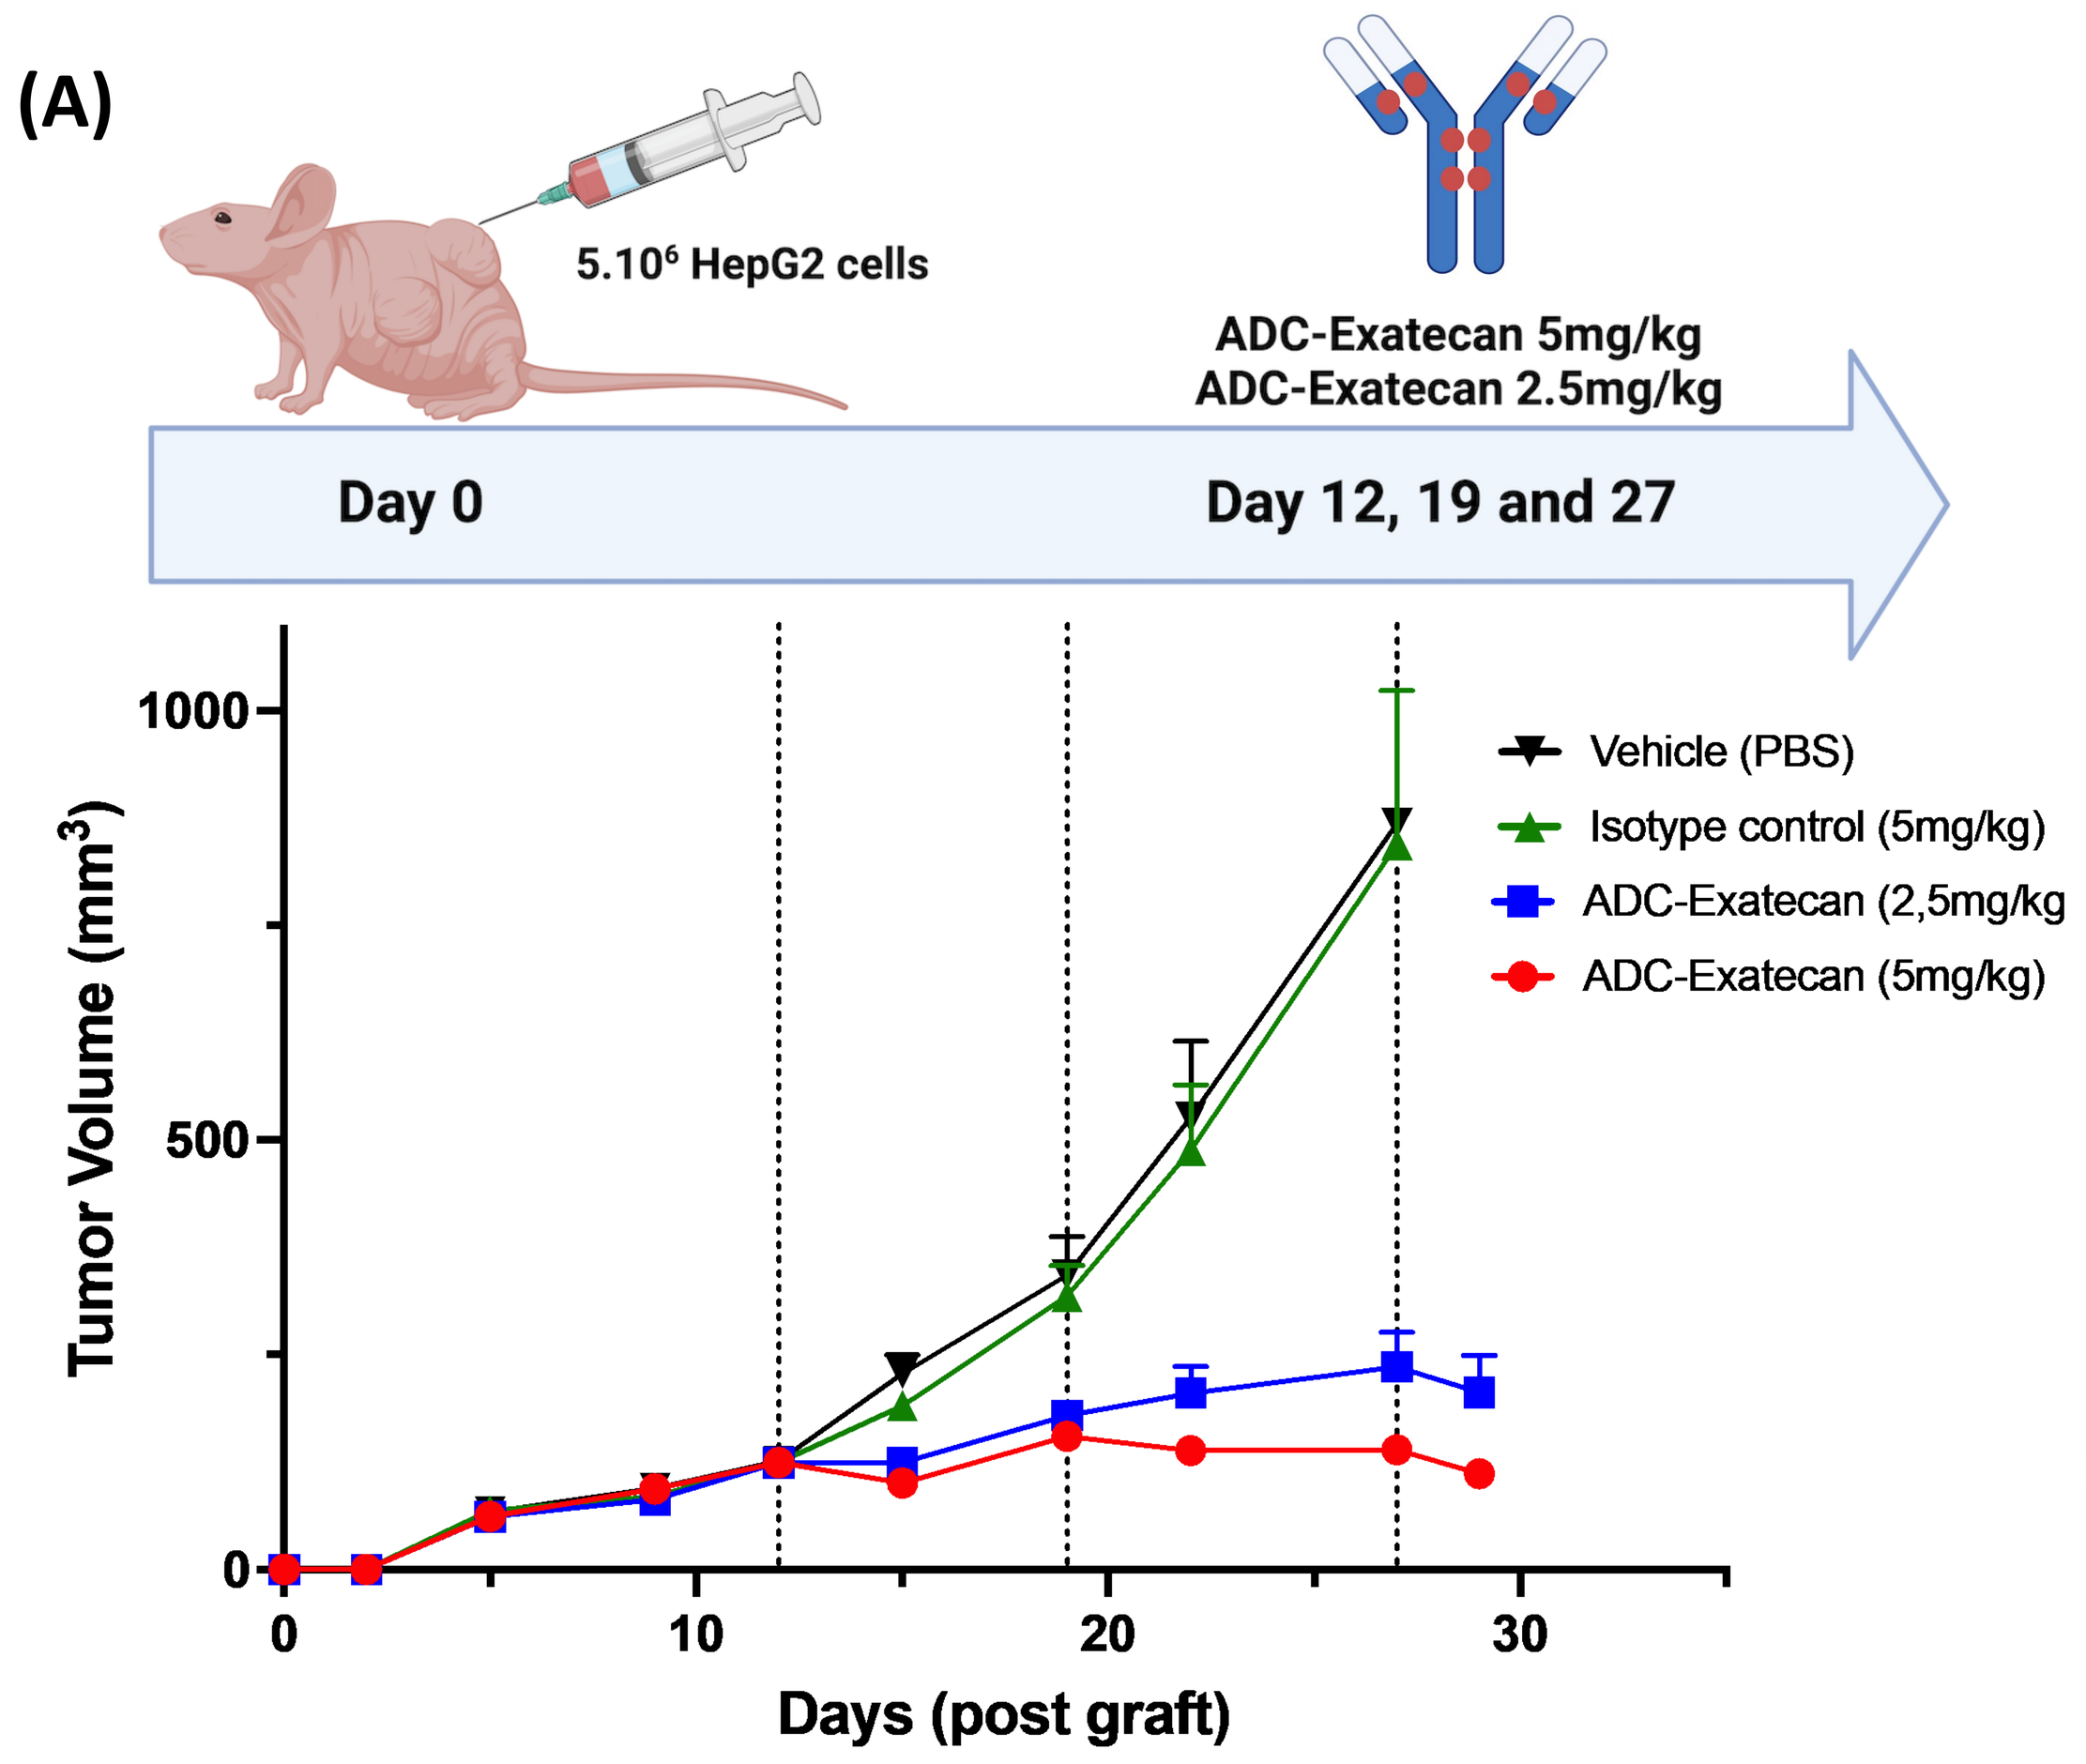

(A)
5.10⁶ HepG2 cells
ADC-Exatecan 5mg/kg
ADC-Exatecan 2.5mg/kg
Day 0
Day 12, 19 and 27
Tumor Volume (mm³)
Days (post graft)
Vehicle (PBS)
Isotype control (5mg/kg)
ADC-Exatecan (2,5mg/kg
ADC-Exatecan (5mg/kg)

Fig.7 (A) Tumor volume of HepG2 cells engrafted subcutaneously into the flank of NMRI nude mice. Vehicule, Isotype control and ADC-Exatecan have been administrated once a week during three consecutive weeks at 2,5 or 5 mg/kg.

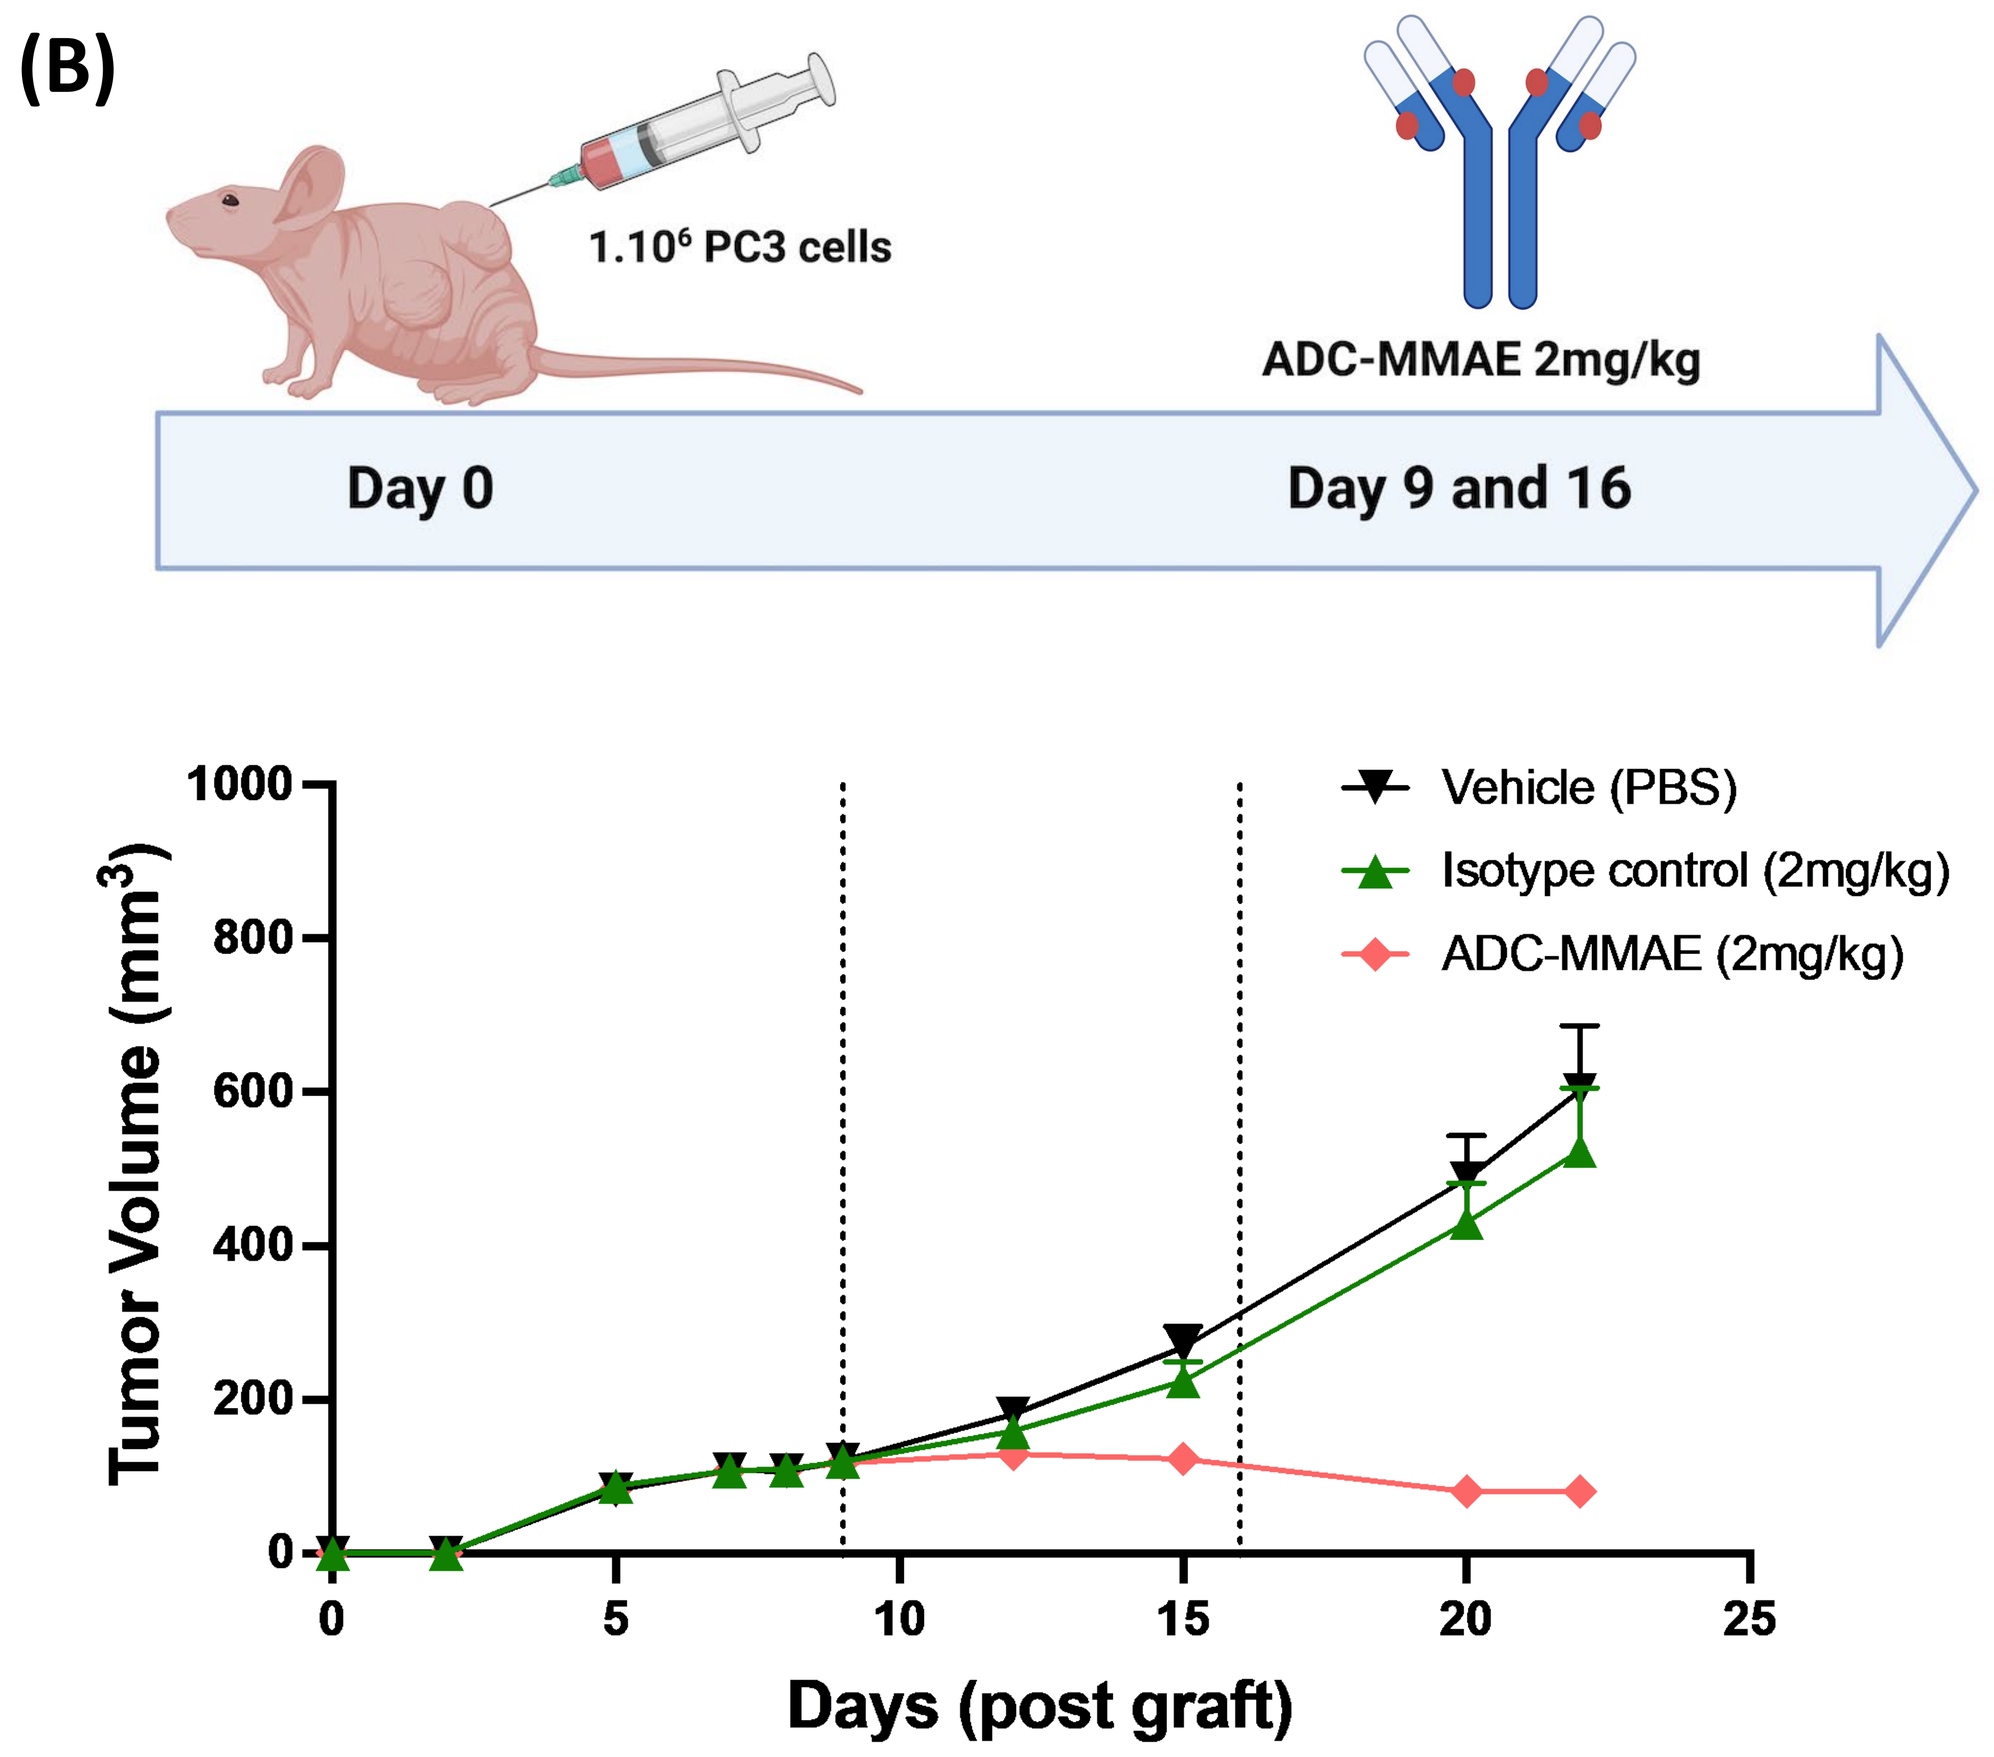

(B)
1.10⁶ PC3 cells
ADC-MMAE 2mg/kg
Day 0
Day 9 and 16
Tumor Volume (mm³)
Days (post graft)
Vehicle (PBS)
Isotype control (2mg/kg)
ADC-MMAE (2mg/kg)

Fig.7 (B) Tumor volume of PC3 cells engrafted subcutaneously into the flank of NMRI nude mice. Vehicule, Isotype control and ADC-MMAE have been administrated once a week during two consecutive weeks at 2mg/kg.

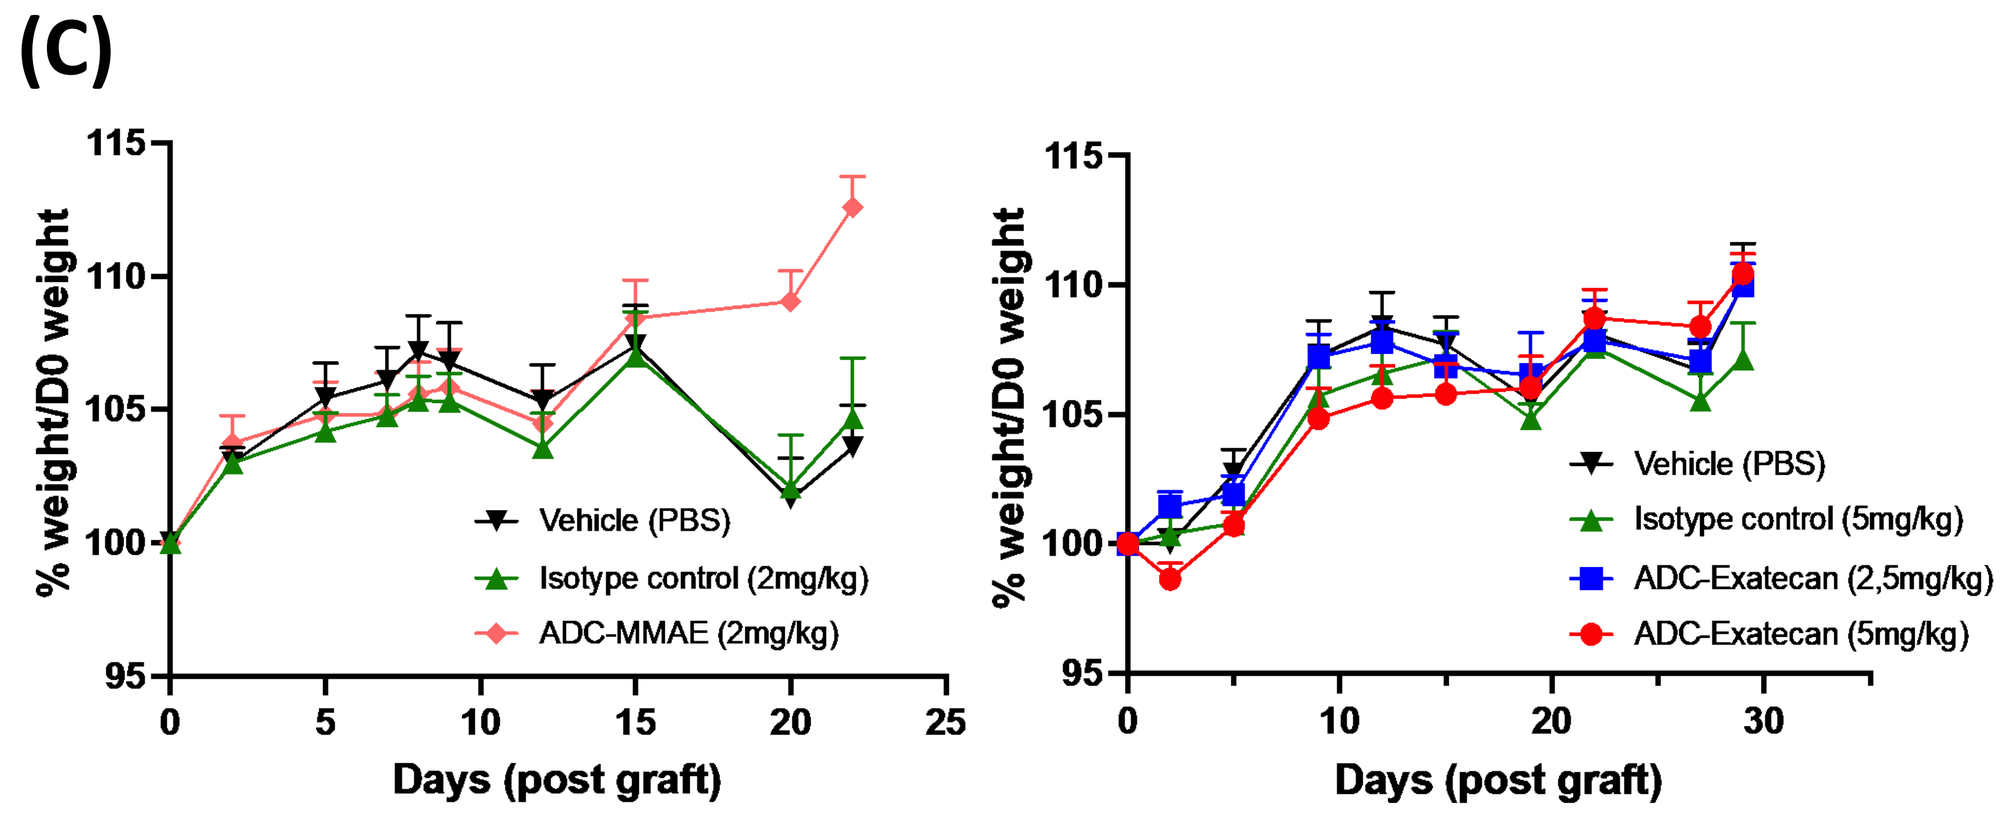

(C)
% weight/D0 weight
Days (post graft)
Vehicle (PBS)
Isotype control (2mg/kg)
ADC-MMAE (2mg/kg)
Vehicle (PBS)
Isotype control (5mg/kg)
ADC-Exatecan (2,5mg/kg)
ADC-Exatecan (5mg/kg)

Fig.7 (C) Percent body weight change calculated relative to animal weight at the start of study shows ADC-Exatecan and ADC-MMAE were well tolerated.

Summary

- DPP4 (CD26) is a cell surface antigen that is overexpressed on multiple solid tumor indications and has been overexpressed during senescence induction by cancer treatment.
- Anti-DPP4 antibody is well internalized by DPP4 positive cancer cells making DPP4 an attractive ADC target.
- STX-1 ADCs exhibit cytotoxic activity against a large panel of DPP4-positive tumor cell lines.
- Consistent with the *in vitro* activity, STX-1 ADCs display efficacy in DPP4+ xenograft models.

STX-1 ADCs represent a promising therapeutic candidate to target cancer and senescent cancer cells

References

1. *Kim KM et al. Identification of senescent cell surface targetable protein DPP4. Genes Dev. 2017 Aug 1;31(15):1529-1534.*
2. *Wang L et al. Exploiting senescence for the treatment of cancer. Nat Rev Cancer. 2022 Jun;22(6):340-355.*
3. *Schmitt CA et al. Senescence and cancer - role and therapeutic opportunities. Nat Rev Clin Oncol. 2022 Oct;19(10):619-636.*
4. *STARKAGE TX Patents Family: #US20240009242/A1, #EP4034561A2 'Senescent cell-associated antigen-binding domains, antibodies and chimeric antigen receptors comprising the same, and uses thereof'.*

AACR Meeting, San Diego (USA), 2024

©2024 StarkAge Therapeutics. All rights reserved.